PIH67

A Review of Patient-Reported Outcome Orphan Drug Labels in the United States From January 2006-September 2013: Analysis of Evidence for Orphan Drug PRO Label Claims

Marci Clark,[1] Chelsea Simons,[2] Emily Haydysch,[3] Carla DeMuro,[3] Ari Gnanasakthy[4]

[1]RTI Health Solutions, Ann Arbor, Michigan, United States;
[2]University of Michigan College of Pharmacy, Ann Arbor, Michigan, United States;
[3]RTI Health Solutions, Research Triangle Park, North Carolina, United States;
[4]Novartis, East Hanover, New Jersey, United States

ABSTRACT

Objectives: Previous reviews of patient-reported outcome (PRO) label claims did not distinguish products with orphan designation. Literature suggests that less evidence is required for orphan drug approvals compared with non-orphan drugs. This study aimed to identify orphan drug products approved by the US Food and Drug Administration (FDA) between 2006 and 2013, the prevalence of PRO label claims for these products, and levels of evidence supporting PRO claim approval.

Methods: Using the Drugs@FDA website, new molecular entities and biologic licensed agents approved between January 2006 and September 2013 with orphan review classification were identified, with subsequent label review. Priority or standard review was also noted. For products with PRO claims, labels and medical and summary review sections from FDA drug approval packages were reviewed to identify indication, reviewing division, PRO endpoint status, PRO measure type, number of phase 3 trials supporting claims, and PRO-related comments from the Study Endpoints and Labeling Development (SEALD) team. Descriptive data were recorded in Microsoft Excel; frequency of measured characteristics was analyzed.

Results: For the 43 products identified, a total of 7 PRO label claims were granted to 5(12%) products. Five orphan products achieved 7 PRO claims. Priority and standard review prevalence were similar (44% vs. 40%, respectively) for orphan products; most products (4 of 5) with PRO claims had priority review. A slight majority (60%; 3 of 5 orphan products) included PRO claims supported by ≤ 1 phase 3 trial. Signs/symptoms measures and secondary PRO endpoints were most common (57% each). FDA reviewing division varied. One product had documented comments from SEALD.

Conclusions: For the time period evaluated, orphan products rarely included a PRO label claim. These claims may not be needed for orphan product differentiation to the degree that they are for non-orphan products. PRO claims achieved for orphan products appear to require less supporting evidence for approval than non-orphan products, consistent with orphan drug approval expectations.

Table 1 Endpoint Status, PRO Measures and Label Claim Language for Orphan Products With Label Claims

Orphan Product/ Approval	Endpoint Status/PRO Measures	Label Claim Language
Jakafi (ruxolitinib) November 16, 2011	**Secondary endpoint:** • Modified Myelofibrosis Symptom Assessment Form (MFSAF) v2.0 diary	• "[P]roportion of patients with a 50% or greater reduction in Total Symptom Score from baseline to Week 24 [was] measured by the modified Myelofibrosis Symptom Assessment Form (MFSAF) v2.0 diary" • "A higher proportion of patients in Jakafi group had 50% or greater reduction in Total Symptom Score vs. placebo group, with a median time to onset of less than 4 weeks" • "All 6 of the symptoms contributed to the higher Total Symptom Score response rate in the group treated with Jakafi"
Firazyr (icatibant) August 25, 2011	**Primary endpoint:** • 3-item composite visual analog score (VAS) comprising averaged assessments of skin swelling, skin pain, and abdominal pain **Secondary endpoints:** • 3 VAS items assessing skin swelling, skin pain, and abdominal pain scored individually • Rescue medication use (PRO instrument not specified in label or DAP; however, a diary was implemented in the trial, which may have captured this endpoint)	• "Response to therapy was primarily assessed using visual analog scores on a 100 mm scale and [patient-reported] symptom scores for abdominal and cutaneous pain and swelling" • "The primary endpoint was assessed using a 3-item composite visual analog score (VAS), comprised of averaged assessments of skin swelling, skin pain, and abdominal pain. Response was defined as at least a 50% reduction from the pretreatment composite 3-item VAS score" • Median time to 50% reduction in symptoms: 2 hours for the Firazyr group; 19.8 hours for the placebo group • Median time for almost complete symptom relief (VAS < 10 mm): 8 hours for the Firazyr group; 36 hours for the placebo group • 7% of Firazyr patients used rescue medication, compared with 40% of placebo patients • "In an assessment of the first 5 FIRAZYR-treated attacks (621 doses for 582 attacks), the median times to a 50% reduction from the pretreatment composite 3-item VAS score were similar across attacks" • Across trials, Firazyr median time to 50% reduction from baseline: 2.0-2.3 hours • "Patients who administered FIRAZYR during an acute attack of [hereditary angioedema] had a median time to 50% reduction from the pretreatment composite 3-item VAS score of 2.6 hours"
Banzel (rufinamide) March 3, 2011	**Primary endpoint:** • Parent/Guardian Global Evaluation of the patient's condition	• Improvement in Seizure Severity Rating from Global Evaluation: 53.4 in the rufinamide group compared with 30.6 in the placebo group (P = 0.0041)
Makena (hydroxyprogesterone caproate) February 3, 2011	**Primary endpoint:** • Ages and Stages Questionnaire (ASQ)	• "The proportion of children whose [ASQ] scores met the screening threshold for developmental delay in each developmental domain was similar for each treatment group"
Letairis (ambrisentan) June 15, 2007	**Secondary endpoint:** • Short Form 36-item Health Survey (SF-36)	• "SF-36 Health Survey [was] assessed" • "LETAIRIS can improve your ability to exercise and it can help slow down the worsening of your physical condition and symptoms"[b]

[a] Primary endpoint in a follow-up safety study evaluating the children whose mothers had participated in the phase 3 trial.
[b] This statement was included in the patient guide for Letairis.
Source: FDA, 2013.[6]

BACKGROUND

- The US FDA orphan drug definition is as follows:[1]
 - Intended to treat a disease or condition that affects fewer than 200,000 people OR will be administered to fewer than 200,000 people per year

 OR
 - Intended to treat a disease or condition that affects 200,000 people or more OR will be administered to 200,000 or more people per year with no reasonable expectation that costs of research and development of the drug for that indication can be recovered by drug sales
- All original new drug applications, original biologic license applications, and efficacy supplements receive a review designation of either "priority" or "standard."
 - Priority review:
 - Intended to direct overall attention and resources to evaluation of drug applications that may provide significant improvements to public health if approved
 - Assigned to applications for drugs that treat serious conditions and provide significant improvements in the safety or effectiveness of the treatment, diagnosis, or prevention of serious conditions compared with available therapies
 - Goal date for FDA action (approval or complete response action letter): within 6 months of receipt
 - Standard review:
 - Applications for drugs that do not meet priority designation criteria
 - Goal date for FDA action (approval or complete response action letter): within 10 months of receipt
- Orphan products receiving priority review status rose from 35% in 2000-2002 to 50% in 2006-2008; share of orphan significant biologics receiving priority review status rose from 17% to 67%.[2]
- Drugs developed for common conditions most often require two phase 3 trials for FDA approval, whereas orphan drugs are more often approved based on a single efficacy trial.[1]
- Therefore, we hypothesized that the level of evidence needed to support a PRO label claim for a drug indicated for a rare disease (i.e., designated as "orphan" status by the FDA) is less substantial than the evidence required to support a PRO label claim for drugs indicated for common conditions.
- Previous reviews of PRO label claims did not distinguish products with orphan designation.[3,4]

OBJECTIVE

- To identify orphan drug products approved by the US FDA between 2006 and 2013, prevalence of PRO label claims for these products, and evidence supporting PRO claim approval.

METHODS

- Using the drug approval reports by month search on the Drugs@FDA website,[5] we identified new molecular entities (NMEs) and biologic licensed agents (BLAs) approved between January 2006 and September 2013 with orphan review classification; standard or priority review classification was also noted for orphan products.
 - Products containing substances previously marketed with a different brand name or set of indications, as a different dosage form or strength, or as a combination product of previously marketed entities were excluded.
- Orphan product labels were reviewed to identify those with and those without PRO label claims.
- For orphan products with PRO label claims:
 - Using the search by drug name on the Drugs@FDA website,[6] we identified and reviewed each product label and drug approval package (DAP), particularly the medical review and summary review sections, to identify and retrieve the following information:
 - PRO label claim language and PRO measure supporting claim
 - Reviewing division
 - Indication
 - PRO endpoint status (primary, secondary, tertiary/exploratory)
 - PRO claim type: symptoms, functioning, health-related quality of life (HRQOL), patient global rating, or "other"; a product label may contain more than one type of claim.
 - Number of phase 3 randomized, controlled trials (RCTs) supporting approval of the PRO claim
 - PRO-related comments from FDA SEALD team
- Microsoft Excel 2010 was used to record descriptive data and analyze the frequency of measured characteristics.

RESULTS

- During the review period, 43 orphan products were identified.
 - Priority and standard review prevalence were similar (44% vs. 40%, respectively) for most orphan products; priority or standard review classification was not specified for 16% of orphan products.
- Of the 43 orphan products, 5 products (12%) were granted PRO label claims (Table 1).
 - For the 5 products with claims, a total of 7 PRO claims were granted.
- As shown in Table 2, signs/symptoms and secondary endpoints most commonly (57% each) supported orphan product PRO claims.

Table 2 Endpoint Status and PRO Claim Type

	Signs/ Symptoms	Functioning	HRQOL	Patient Global Rating	Other	Total
Primary endpoint	n = 1 (14%)	n = 0 (0%)	n = 0 (0%)	n = 1 (14%)	n = 1[a] (14%)	n = 3 (43%)
Secondary endpoint	n = 3 (43%)	n = 0 (0%)	n = 1 (14%)	n = 0 (0%)	n = 0 (0%)	n = 4 (57%)
Total	n = 4 (57%)	n = 0 (0%)	n = 1 (14%)	n = 1 (14%)	n = 1[a] (14%)	N = 7 (100%)

[a] PRO claim based on the caregiver-reported and completed Ages and Stages Questionnaire.

- Of the 5 orphan drug products with PRO claims, 3 products (60%) had a PRO as a primary endpoint.
 - PRO claims were based on data from one or more phase 3 RCTs for Firazyr and Banzel; a single safety study provided the basis for Makena's PRO claim.
 - The primary endpoint claim types were signs/symptoms (1), global rating (1), and other (1).
- Table 3 provides an overview of the review classification, reviewing division, and level of phase 3 support for orphan products with PRO claims.
 - Most orphan products with PRO claims (4 of 5; 80%) received priority review by the FDA.
 - FDA reviewing division varied for orphan products with PRO claims.
 - A slight majority (3 of 5; 60%) of orphan products included PRO claims supported by no more than one phase 3 trial.

Table 3 Review Classification, Reviewing Division, and Level of Phase 3 Support

Orphan Product/ Approval	Review Classification (Priority/ Standard)	Reviewing Division	Number of Phase 3 RCTs Supporting PRO Claims
Jakafi (ruxolitinib) November 16, 2011	Priority	Division of Hematology Products	1
Firazyr (icatibant) August 25, 2011	Priority	Division of Pulmonary, Allergy and Rheumatology Products	3
Banzel (rufinamide) March 3, 2011	Standard	Division of Neurology Products	1
Makena (hydroxyprogesterone caproate) February 3, 2011	Priority	Division of Reproductive and Urologic Products	0[a]
Letairis (ambrisentan) June 15, 2007	Priority	Division of Cardiovascular and Renal Products	2

[a] A single follow-up safety study evaluating the children whose mothers had participated in the phase 3 trial supported the PRO claim.

- FDA SEALD comments were identified in the Medical Review section of the DAP for 1 of 5 (20%) orphan products with PRO claims and were provided in advance of the sponsor completing the final phase 3 RCT to support approval of the product (Firazyr):
 - The verbal descriptor scale (VDS) (used in the observational nonintervention study to define minimal clinically significant difference for the VAS with the initial new drug application) required more rigorous definition. Incorporation of a temporal element in the VDS made this measure less reliable.
 - The patient population in the PRO validation study was not representative of the patients participating in the phase 3 study.
 - Patient diaries used in phase 3 studies should have been included in the validation protocol.
 - There are potential labeling limitations for claims supported solely by a VAS.

DISCUSSION

- The following was observed among all NMEs/BLAs with orphan review classification that were approved by the FDA between January 2006 and September 2013:
 - These products rarely included a PRO label claim. Because of the rarity of the diseases for which orphan drugs are typically approved, and possibly because these drugs may have fewer competitors, achieving PRO claims (when not required as a primary endpoint for drug approval) may not be as important for product differentiation as it is for products approved for more common conditions with multiple treatment competitors.
 - These products received priority review slightly less often (44%) than was reported by Tufts Center for the Study of Drug Development[2] for all orphan products for the period of 2006-2008 (50%).
- The following was observed among all NMEs/BLAs with orphan review classification that were approved by the FDA between January 2006 and September 2013 and that achieved PRO claims:
 - These products may require less clinical trial evidence for approval than non-orphan products: 60% of approved orphan drugs in this time period required 1 or fewer phase 3 trials for PRO claim approval.
 - These products received priority review in the majority (80%) of cases.
- In comparison with previous study results reported by Gnanasakthy and colleagues[3] on PRO label claims in NMEs/BLAs approved between January 2006 and December 2010 for all conditions (both common and rare):
 - Results of this study were similar in that symptom-related claims were the most commonly reported type of PRO claim.
 - Results of this study were different in the following ways:
 - No functioning-related claims were identified in our findings, whereas 25% of claims identified by Gnanasakthy and colleagues[3] were for functioning.
 - Primary versus nonprimary endpoint status rate differed:
 - Gnanasakthy and colleagues[3] found that 20 of 28 (71%) PRO label claims were supported by primary endpoints; 29% were supported by nonprimary endpoints.
 - The current study found that 3 of 7 (43%) PRO label claims were supported by primary endpoints; 57% were supported by nonprimary endpoints.
 - PRO instrument type for primary endpoints differed:
 - Gnanasakthy and colleagues[3] found that all primary endpoints supporting PRO claims were based on signs/symptoms.
 - The current study found that only 1 of 3 primary endpoints supporting PRO claims for orphan products reviewed were based on signs/symptoms.
- Documented comments show that SEALD appears to have influenced the Division of Pulmonary, Allergy and Rheumatology Products to require additional documentation of content validity and further validation of the patient diary used to collect the information for the primary PRO endpoint prior to approval of Firazyr, consistent with the FDA PRO guidance requirements.[7]

CONCLUSIONS

- Drug manufacturers may not consider PRO claims to be key for product differentiation.
- PRO claims granted for NMEs/BLAs with orphan review classification and approved by the FDA between January 2006 and September 2013 appear to have required fewer phase 3 RCTs for approval than non-orphan products, consistent with orphan drug approval expectations.

LIMITATIONS

- A small sample of orphan products with PRO label claims was evaluated (N = 5).
- The evidence was limited to published information and aspects of DAPs made available by the FDA.
- The evidence was limited to product review classification designated in the Drugs@FDA database:
 - Some drugs may have been missing orphan review designation in the database.
 - Seven orphan products (16%) had unspecified standard or priority review classification.

REFERENCES

1. Institute of Medicine Committee on Accelerating Rare Diseases Research and Orphan Product Development; Field MJ, Boat TF, editors. Rare diseases and orphan products: accelerating research and development. Washington (DC): National Academies Press (US); 2010. 3, Regulatory Framework for Drugs for Rare Diseases. Available at: http://www.ncbi.nlm.nih.gov/books/NBK56185/. Accessed October 10, 2013.
2. Tufts Center for the Study of Drug Development. U.S. orphan product designations more than doubled from 2000-02 to 2006-08. Impact Report. 2010;12(1). Available at: http://csdd.tufts.edu/files/uploads/2010_jan-feb_summary.pdf. Accessed October 10, 2013.
3. Gnanasakthy A, Mordin M, Clark M, DeMuro C, Fehnel S, Copley-Merriman C. A review of patient-reported outcome labels in the United States: 2006 to 2010. Value Health. 2012 May;15(3):437-42.
4. DeMuro C, Clark M, Doward L, Evans E, Mordin M, Gnanasakthy A. Assessment of PRO label claims granted by the FDA as compared to the EMA (2006-2010). Value Health. 2013 Dec;16(8):1150-5.
5. Food and Drug Administration. Drug approval reports by month. Available at: http://www.accessdata.fda.gov/scripts/cder/drugsatfda/index.cfm. Accessed October 10, 2013.
6. Food and Drug Administration. Search by drug name. Available at: http://www.accessdata.fda.gov/scripts/cder/drugsatfda/index.cfm. Accessed October 15, 2013.
7. Food and Drug Administration. US Department of Health and Human Services. Guidance for industry. Patient-reported outcome measures: use in medical product development to support labeling claims. December 2009. Available at: http://www.fda.gov/downloads/Drugs/GuidanceComplianceRegulatoryInformation/Guidances/UCM193282.pdf. Accessed March 11, 2014.

CONTACT INFORMATION

Marci Clark, PharmD
Director, Patient-Reported Outcomes

RTI Health Solutions
3005 Boardwalk St., Suite 105
Ann Arbor, MI 48108

Phone: +1.734.213.5638
Fax: +1.734.213.6169
E-mail: mclark@rti.org

Presented at ISPOR 19th Annual International Meeting; May 31-June 4, 2014; Montreal, QC, Canada