Development and Validation of an HPLC Method with UV Detection to Support the Clinical Development of Nepicastat for the Treatment of Cocaine Dependency

T. Jester[1], J. Hines[1], A. Gilbert[1], A. Vu[1], M. Park[2]
[1]RTI International, 3040 Cornwallis Road, Research Triangle Park, NC 27709, USA
[2]Chemistry and Pharmaceutics Branch, DPMC, National Institute on Drug Abuse, Bethesda, MD 20892

Purpose

Nepicastat, 5-(aminomethyl)-1-[(2S)-5,7-difluoro-1,2,3,4-tetrahydronaphthalen-2-yl]-1,3-dihydro-2H-imidazole-2-thione hydrochloride is a potent and selective inhibitor of the enzyme dopamine β-hydroxylase (DBH). The compound has demonstrated potential as a treatment for cocaine dependence and post-traumatic stress disorder (PTSD). Under a Collaborative Research and Development Agreement (CRADA), the National Institute on Drug Abuse (NIDA) and Biotie Therapies will evaluate the safety and efficacy of nepicastat in the treatment of cocaine dependence. To support the clinical development a reverse-phase HPLC-UV method was developed and validated for the accurate quantitation and impurity profiling of the drug substance.

Description of Compound Nepicastat hydrochloride

$C_{14}H_{15}N_3F_2S \cdot HCl$

Nominal Mol. Wt. 331.81

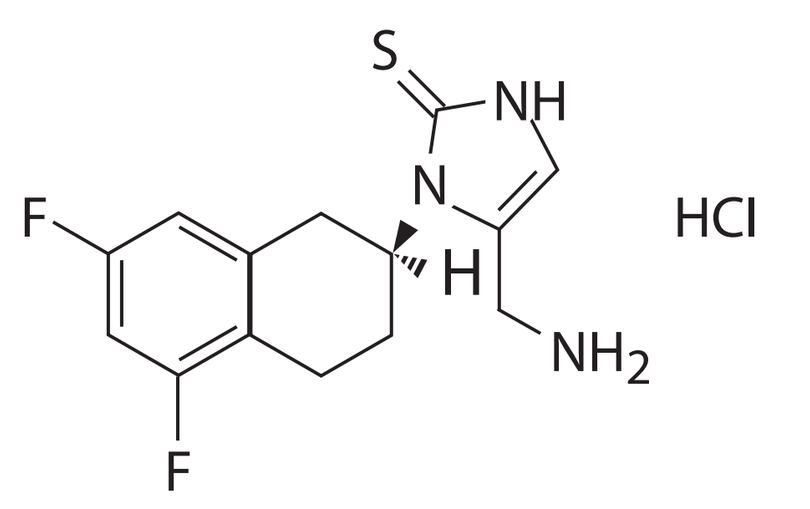
S
NH
N
F
H
HCl
NH2
F

Methods

Test sample aliquots were dissolved in 25% acetonitrile/75% phosphate buffer (v/v) and analyzed by HPLC-UV. The reverse-phase chromatographic separation was performed using a Waters Sunfire C18 column (150 x 4.6 mm, 3.5 µm particles) and UV detection (263 nm) with acetonitrile/water mobile phases containing 0.1% (v/v) trifluoroacetic acid.

HPLC Conditions:

Equipment:	HPLC systems (Waters) equipped with an autosampler, column-temperature-controller, photodioade array or dual wavelength detector and Waters chromatographic data system
Mode:	Reverse phase, isocratic
Column:	Waters Sunfire C18, 4.6 x 150 mm id, 3.5 µm
Column Temp.:	30°C
Mobile Phase:	24% Acetonitrile:76% Water containing 0.1% TFA (v/v)
Flow Rate:	1.0 mL/minute
Detection:	UV at 263 nm
Sample Diluent:	75:25 (v:v) Potassium Phosphate Buffer:Acetonitrile
Injection Volume:	10 µL
Run Time:	15 minutes
Assay Concentration:	0.2 mg/mL (nominal)

Representative HPLC assay chromatogram of 0.20 mg/mL Nepicastat drug substance in water

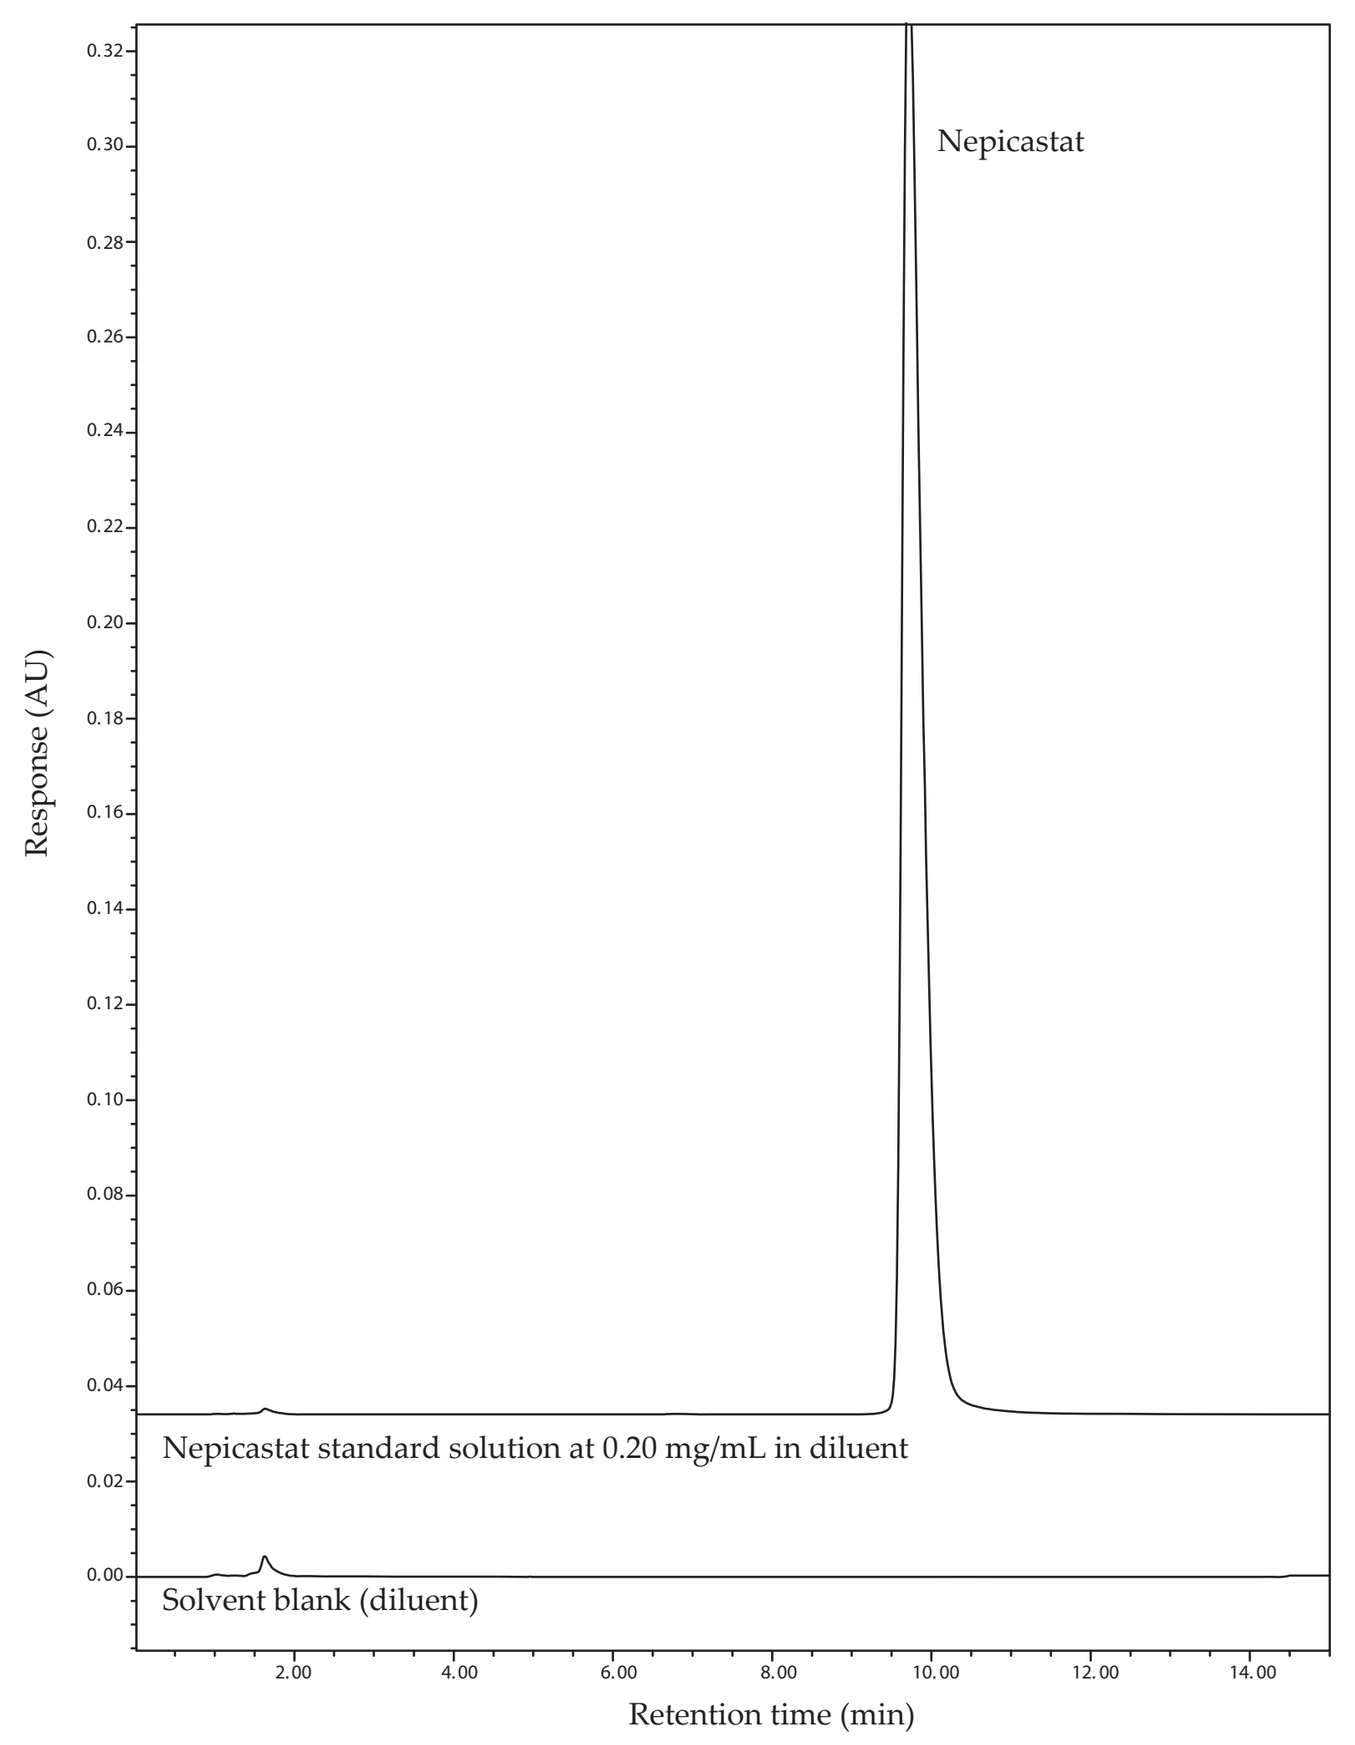
Nepicastat
Nepicastat standard solution at 0.20 mg/mL in diluent
Solvent blank (diluent)
Response (AU)
Retention time (min)

Results

Linearity (y = a + bx) was demonstrated over the range 0.10-0.30 mg/mL (50-150% of assay concentration), with a correlation coefficient (r) of ≥ 0.999 and a y-intercept -2.0 % of the nominal assay level. The method precision for multiple prepared samples was 0.6% (RSD) and the accuracy (recovery) between 99.2% and 100.9% over the linearity range. Specificity was demonstrated by the resolution of impurities and forced decomposition products using photodiode array peak purity. Ruggedness and reproducibility were evaluated across three days, two analysts, two analytical columns, and two HPLC systems. Robustness was demonstrated by confirming method performance with slight changes in flow rate, column temperature, capacity factor (k'), and concentration of TFA in the mobile phase. Sample solution stability was confirmed up to 5 days at refrigerated and room temperature storage.

Linearity:

Linearity was evaluated by analysis of standard solutions containing nepicastat dissolved in 25% acetonitrile:75% phosphate buffer (v:v). Five standard concentrations ranging from 0.101 to 0.302 mg/mL were prepared and analyzed (single injection). The concentration and the peak area response of drug were subjected to regression analysis for calculating the calibration equation and correlation coefficient. Individual back-calculated concentrations for all 5 levels of standards were between 99.1 and 101.2% of target. Acceptance criteria with respect to linearity were met.

Results for Determination of Linearity for the Nepicastat Drug Substance Assay Method

Assay Level (%)	Actual Conc. (mg/mL)	Analyte Response (peak area)	Calc Conc. (mg/mL)	% Error from Actual Conc.
50	0.101	2613771	0.103	1.2
80	0.161	4165576	0.161	-0.24
100	0.202	5217301	0.200	-0.90
120	0.242	6321803	0.242	-0.12
150	0.302	7957738	0.303	0.41

Calculated from regression line: Y = a + bX
a = -105407.8
b = 26590500
$r^2 = 0.9997$
Intercept = -2.0% of the target concentration

Linearity for HPLC/UV Detection of Nepicastat over the Assay Concentration range 0.101–0.302 mg/mL

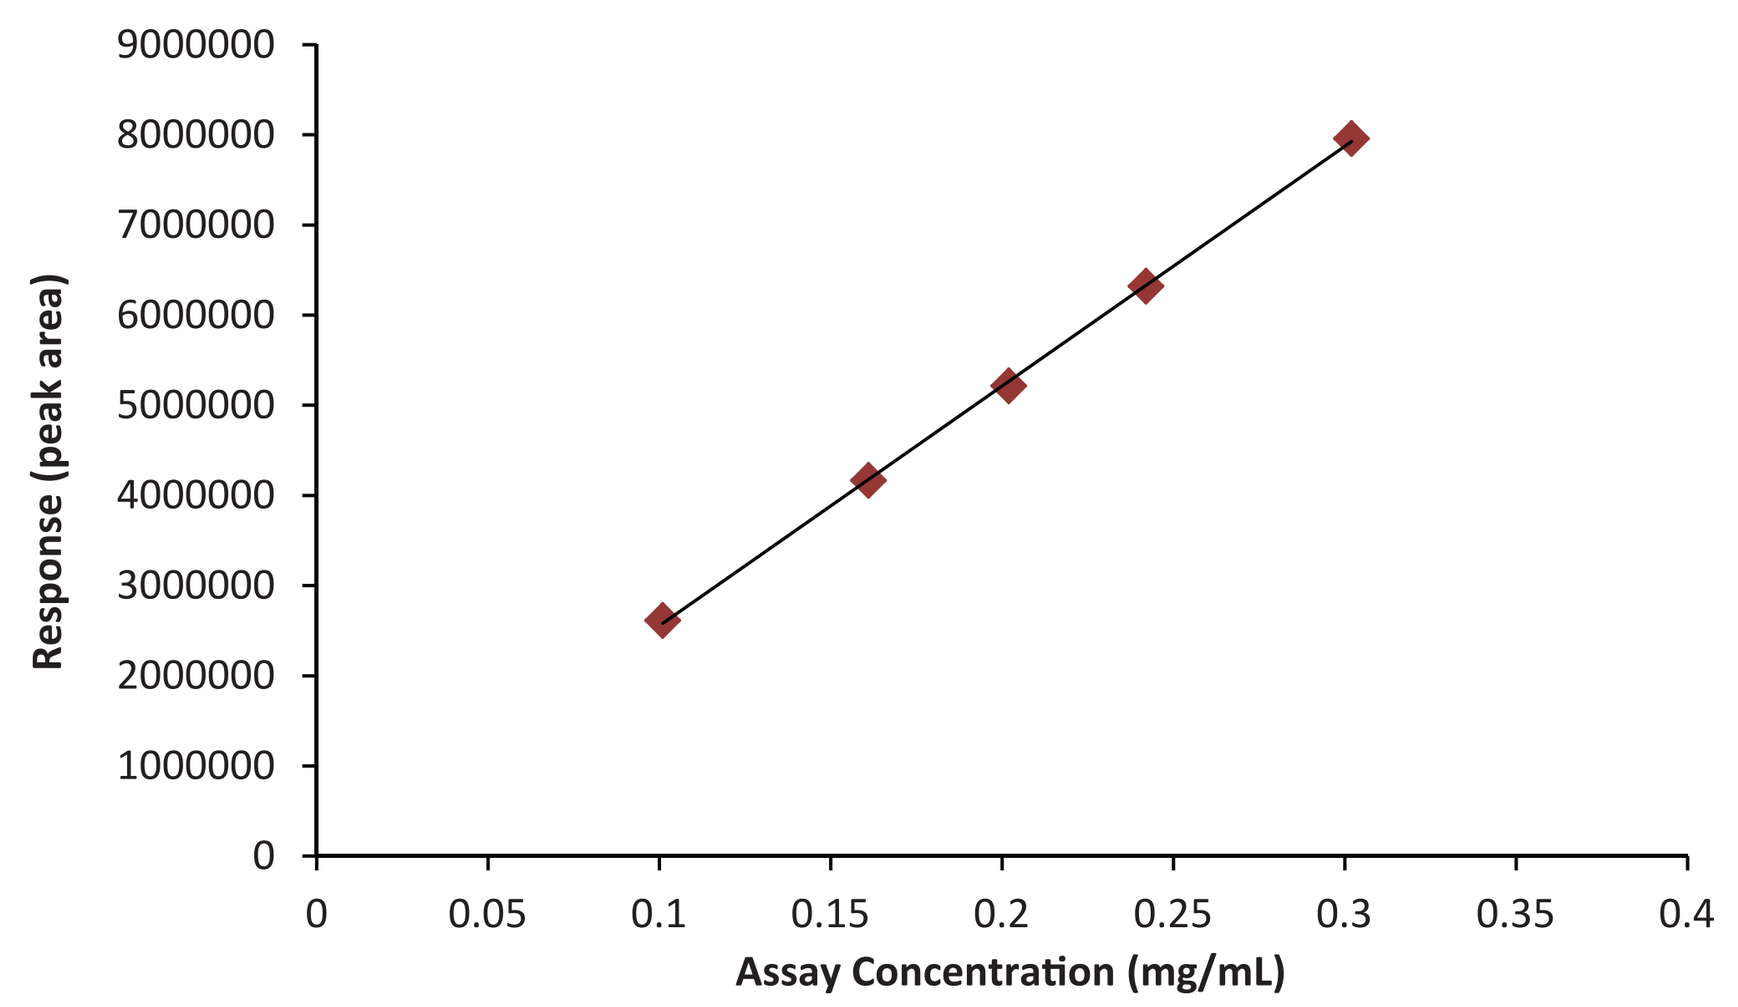
Response (peak area)
Assay Concentration (mg/mL)

Precision (Repeatability):

Method repeatability was determined from analysis of nine standard solutions containing nepicastat, individually prepared, covering the range of the procedure (3 concentrations/3 replicates each). The concentration levels evaluated were at 80%, 100% and 120% of the nominal drug substance assay concentration of 0.20 mg/mL. Acceptance criteria with respect to precision were met.

Results for Determination of Method Repeatability for Nepicastat Drug Substance Assay Method

Assay Level (%)	Replicate	Analyte Retention (min)	RF[a]	Mean ± SD Retention (Min)	%RSD of Retention	Mean ± SD RF	%RSD of RF
80	1	9.742	26265630	9.745 ± 0.003	0.03	26182457 ± 204844.4	0.78
	2	9.746	26332633				
	3	9.747	25949108				
100	1	9.744	26001247	9.743 ± 0.001	0.01	26036822 ± 33368.8	0.13
	2	9.743	26067427				
	3	9.741	26041793				
120	1	9.739	26101115	9.745 ± 0.006	0.06	25944941 ± 254188.1	0.98
	2	9.744	25651636				
	3	9.750	26082072				

[a]analyte response (area)/ analyte concentration

Accuracy:

Assay Accuracy was determined by quantitation of the assay precision samples using the calibration curve calculated from the linearity solutions. Accuracy was measured as the deviation of test results from actual values. Acceptance criteria with respect to accuracy were met.

Results for Determination of Accuracy for Nepicastat Drug Substance Assay Method

Assay Level (%)	Replicate	Actual Conc. (mg/mL)	Calc. Conc. (mg/mL)	Percent (%) Recovery	Mean Recovery (%)
80	1	0.161	0.163	101.2	100.9
	2	0.160	0.162	101.5	
	3	0.163	0.163	100.0	
100	1	0.206	0.205	99.71	99.8
	2	0.207	0.207	99.95	
	3	0.206	0.206	99.86	
120	1	0.247	0.246	99.76	99.2
	2	0.244	0.239	98.09	
	3	0.243	0.242	99.72	

Ruggedness (Intermediate Precision):

Method ruggedness was evaluated across three days, two analysts, two analytical columns, and two HPLC systems to establish the effects of typical variations on the precision of the analytical procedure. The ruggedness (as %RSD) obtained for the analyte chromatographic retention time and analyte response factor (RF) in the nepicastat assay were found to be within the acceptable criteria of %RSD ≤1 for retention at the target assay concentration and %RSD ≤ 2 for RF across variations in days, analysts and equipment.

Results for Determination of Method Ruggedness for Nepicastat Drug Substance Assay Method

Day	Assay Level (%)	Retention Time (min)		RF[a]	
		Average ± SD (n=3)	%RSD	Average ± SD (n=3)	%RSD
1	80	8.903 ± 0.005	0.056	25153647 ± 36409.0	0.14
	100	8.878 ± 0.003	0.034	25075442 ± 225997.6	0.90
	120	8.854 ± 0.004	0.045	25367814 ± 116007.9	0.46
2	80	10.665 ± 0.012	0.113	25075818 ± 46269.2	0.18
	100	10.632 ± 0.013	0.122	24993023 ± 29392.0	0.12
	120	10.598 ± 0.008	0.075	24898224 ± 223584.3	0.90
3	80	8.987 ± 0.004	0.045	24564915 ± 30357.0	0.12
	100	8.959 ± 0.009	0.100	24702868 ± 53806.5	0.22
	120	8.931 ± 0.009	0.101	24644279 ± 26006.5	0.11

[a]analyte response (area)/ analyte concentration

Robustness:

The robustness of the nepicastat method was determined by examining the effect of slight variations on the method performance; specifically, the effect of changes in flow rate, the effect of changes in column temperature, the effect of changes on the capacity factor (k'), and the effect of changes in TFA in the mobile phase. Robustness was assessed by evaluation of system suitability. Results from all robustness tests met all system suitability criteria.

Results for Determination of Method Robustness for Nepicastat Drug Substance Assay Method

Testing		Results			
Parameter	Values	k´	N	T	%RSD for Area
Flow Rate	0.9 mL/min	4.59	11129	1.64	0.3
	1.1 mL/min	4.58	10480	1.56	0.1
Column Temperature	27 °C	5.30	11172	1.63	0.2
	33 °C	4.73	10964	1.59	0.2
Capacity Factor	~4	3.45	10374	1.44	0.2
	~7	6.73	11640	1.39	0.3
Concentration of TFA in the mobile phase	0.09%	4.40	10604	1.48	0.2
	0.11%	4.82	11295	1.46	0.2

Acceptance criteria: k´ = 4-7
N = >2000
T = 0.5 – 2.0
%RSD for area = ≤ 2.0% (n=5)

Specificity:

Accelerated degradation studies were performed to provide an indication of the stability of the drug and specificity of the method. Nepicastat drug substance was stored dry (10 mg) and stressed in solution (10 mg of drug to 10.0-mL solvent) under conditions that usually cause degradation, including: light, solution (MeCN), heat (90 °C), acid (1N HCl), base (1N NaOH) and oxidation (3% H_2O_2). Controls containing no analyte were also exposed with the samples for assessment of chromatographic background.

After 24 h of exposure to the conditions, the solutions were allowed to cool to room temperature and neutralized with acid or base (where necessary). The solutions were brought to volume with sample diluent and aliquoted for analysis. The final concentrations for all solutions were ~0.20 mg/mL nepicastat. Single wavelength data at 263 nm was collected following the method to determine the amount of degradation. The amount of degradation was assessed by comparison of test sample peak area to that of a reference standard solution. Additional PDA detector data was collected for the purposes of the peak purity evaluation (spectrum matching).

Specificity of Nepicastat Drug Substance Assay Method

Sample Condition	Purity Angle (PA)[a]	Purity Threshold (PTHR)[b]	Match Angle (MA)[c]	Match Threshold (MTHR)[d]
Working standard	0.046	0.248	0.000	1.015
	0.043	0.243	0.014	1.014
	0.042	0.250	0.013	1.015
	0.042	0.245	0.017	1.014
	0.043	0.248	0.020	1.015
	0.048	0.246	0.020	1.014
Dry storage, ambient, dark[e]	0.043	0.245	0.041	1.015
MeCN, ambient, light[e]	0.040	0.244	0.021	1.015
MeCN, ambient, dark[e]	0.041	0.242	0.030	1.014
Dry storage, 90 °C, dark[e]	0.042	0.247	0.044	1.015
1N NaOH, ambient, dark[f]	0.049	0.257	0.033	1.018
1N HCl, ambient, dark[e]	0.085	0.341	0.039	1.046
3% H_2O_2, ambient, dark[g]	8.540	12.793	3.134	7.175

[a] spectral homogeneity
[b] sum of noise angle and solvent angle
[c] spectral contrast angle between two spectra
[d] sum of match noise angle and solvent angle
[e] <5% degradation
[f] 5% degradation
[g] >95% degradation

Representative UV Spectra Overlayed to Demonstrate Chromatographic Peak Homogeneity for the Nepicastat Peak after Degradation under Basic pH Conditions (NaOH) used for the Evaluation of Specificity

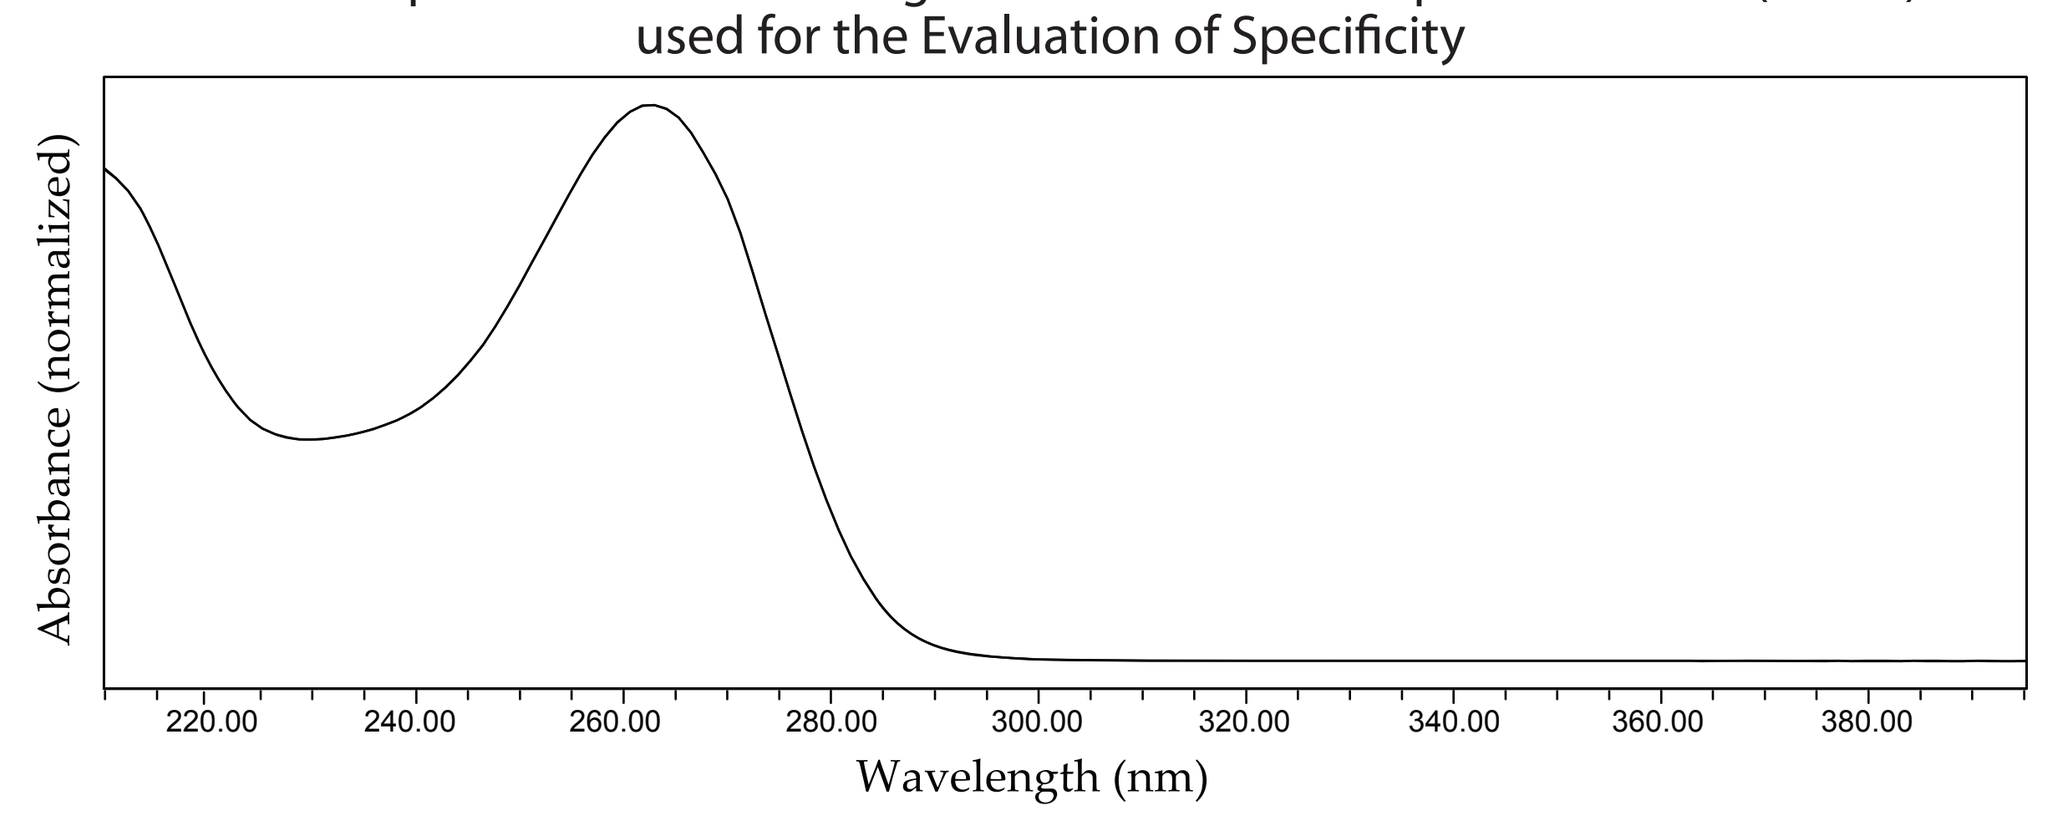
Absorbance (normalized)
Wavelength (nm)

(a) Spectrum taken at the analyte peak apex (8.83 min).
(b) Spectrum taken at the front edge of the analyte peak (8.77 min).
(c) Spectrum taken at the trailing edge of the analyte peak (8.93 min).

Solution Stability:

The stability of the nepicastat drug substance in sample solutions prepared for HPLC assay was evaluated up to 5-days by setting aside an aliquot of the sample solution under ambient laboratory conditions of temperature, humidity, and light (overhead fluorescent) and a second aliquoted stored refrigerated (2-8 °C).
After storage for 5 d, an aliquot from each of the two storage locations was analyzed (triplicate injections) vs. a fresh sample, and the % recovery was calculated for each. The % recovery obtained for the solutions stored for 5 days met acceptance criteria (% Recovery = 100 ± 2%).

Nepicastat Related Impurities:

Nepicastat related organic impurities are determined by a separate gradient extension of the nepicastat assay method. This method was qualified by demonstrating linearity over a range of 0.0402 to 0.301 mg/mL (r^2 = 0.997) and establishing a detection limit of 0.05% assay concentration with a S:N of 3.0.

Nepicastat Drug Substance Impurities Method Summary

Column:	Sunfire C18, 150 x 4.6 mm, 3.5 µm particle size (Waters or equivalent)
Column temperature:	30°C
Detector:	UV at 263 nm
Flow rate:	1.0 mL/minute
Injection volume:	10 µL
Mobile phase A:	0.1% (v/v) TFA in water
Mobile phase B:	0.1% (v/v) TFA in acetonitrile
Gradient Program:	See below
Blank (diluent):	75:25 (v:v) Potassium Phosphate Buffer:Acetonitrile
Sample:	~0.2 mg/mL in sample diluent
Standard:	~0.2 mg/mL in sample diluent

Time	Mobile Phase A (%)	Mobile Phase B (%)
0.0	95	5
30.0	0	100
32.0	0	100
33.0	95	5
43.0	95	5

Representative Nepicastat Drug Substance Impurities Chromatogram

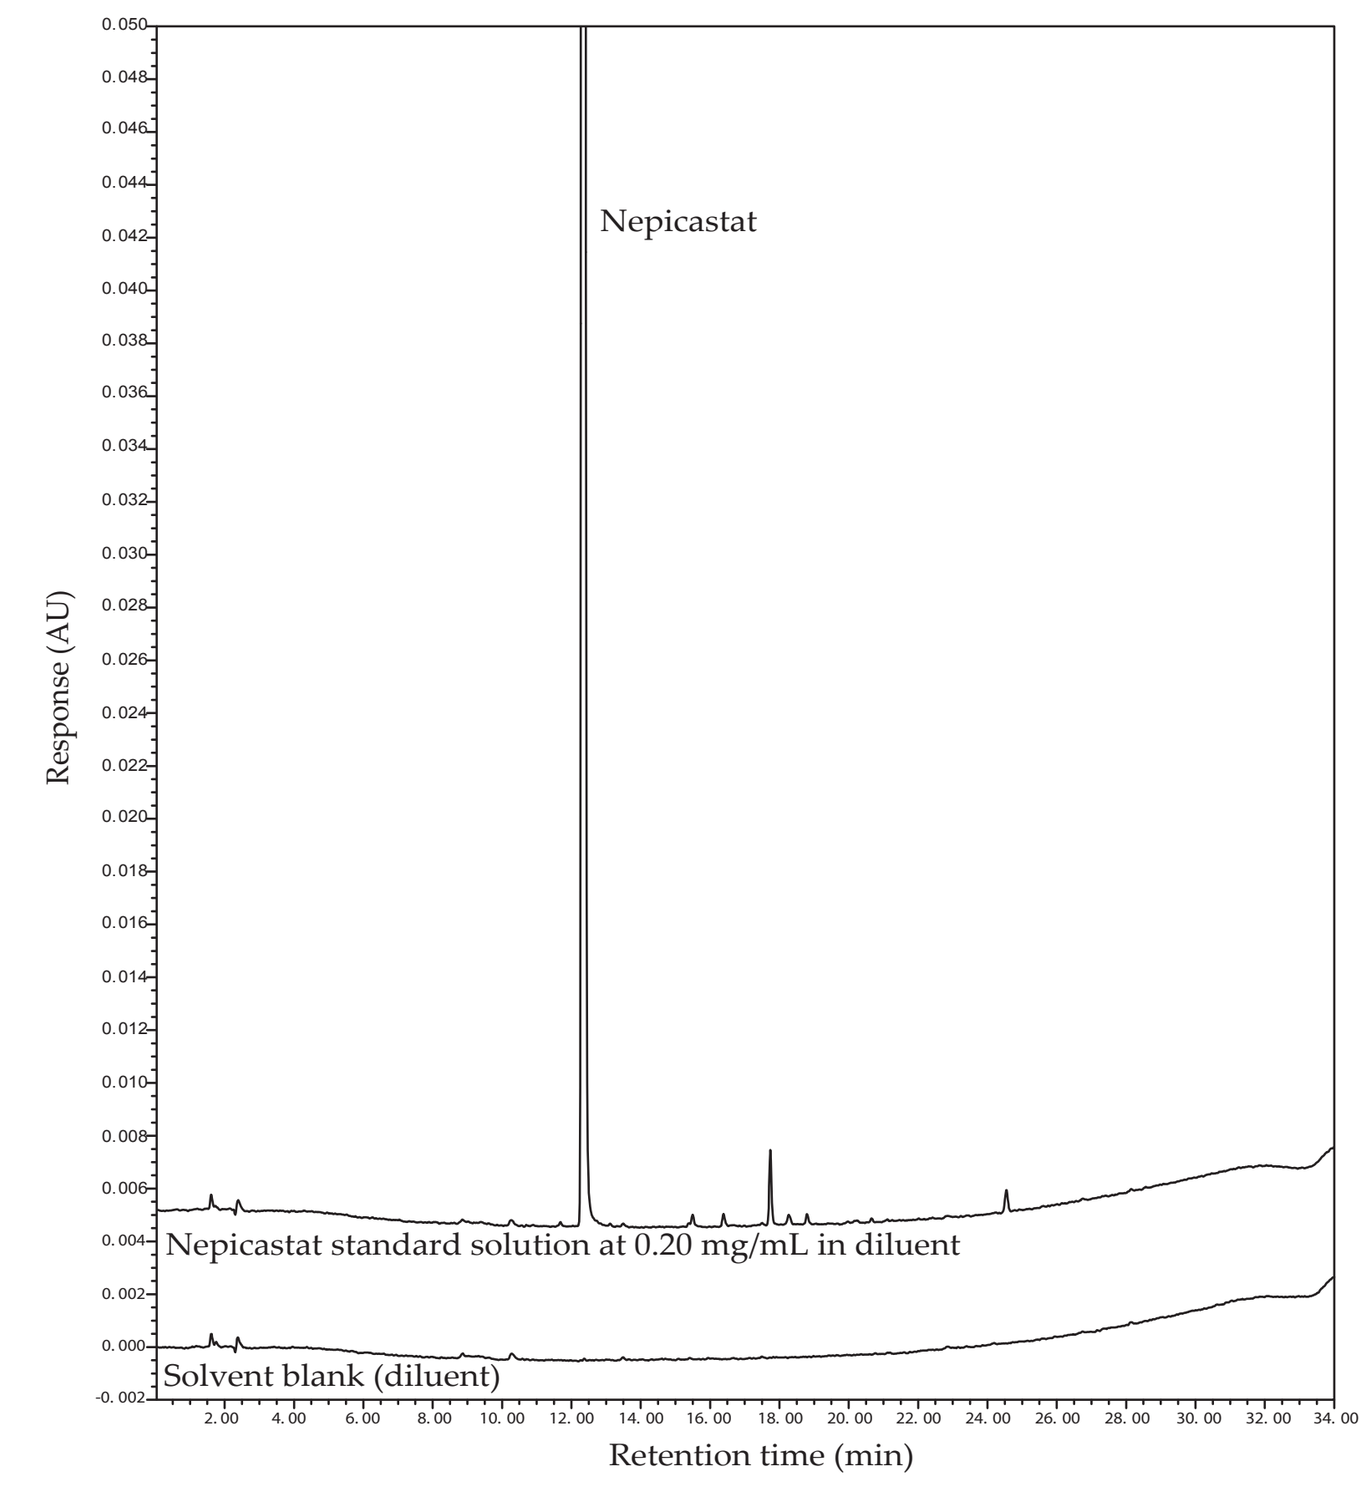
Nepicastat
Nepicastat standard solution at 0.20 mg/mL in diluent
Solvent blank (diluent)
Response (AU)
Retention time (min)

Nepicastat Drug Substance Stability:

The described methods for analysis of nepicastat and related impurities have been used successfully at RTI to evaluate the stability of a cGMP batch of nepicastat drug substance under ICH conditions of heat and humidity for drug substances intended for storage in a freezer including accelerated (40°C/75% RH), intermediate (25°C/60% RH), intermediate (5°C) and long-term (-20°C) stability evaluations.

Trend Chart for Assay of Nepicastat Drug Substance Under ICH Stability Conditions (Intended for Storage in a Freezer)

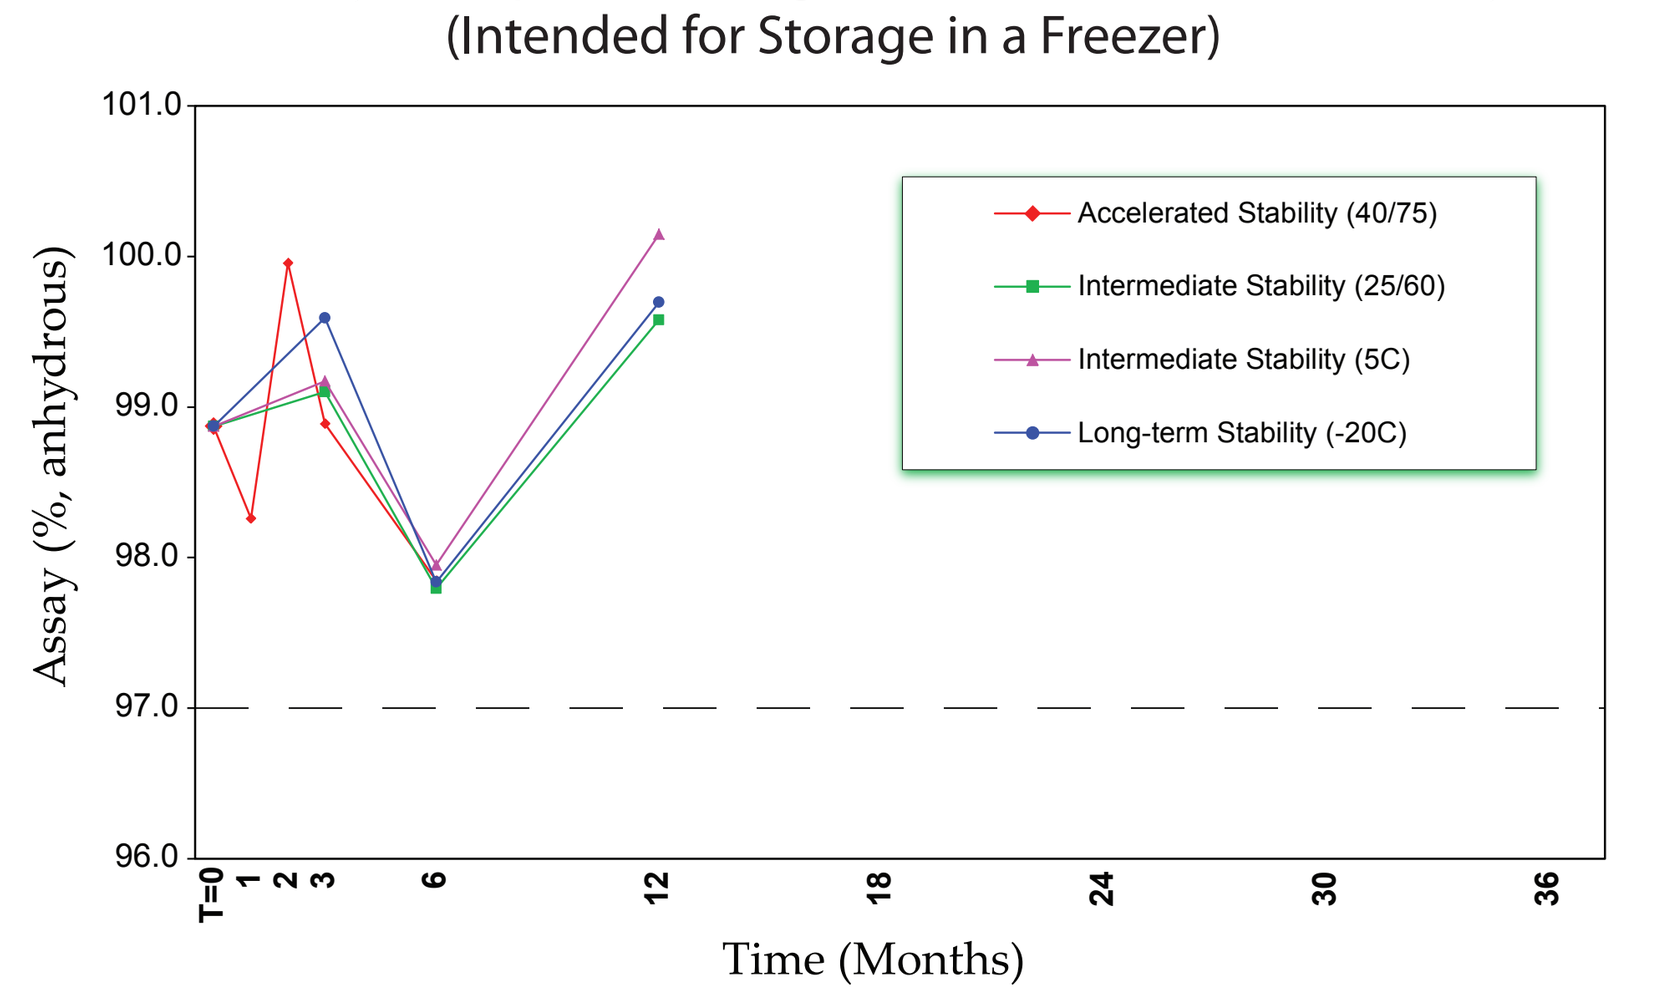
Accelerated Stability (40/75)
Intermediate Stability (25/60)
Intermediate Stability (5C)
Long-term Stability (-20C)
Assay (%, anhydrous)
Time (Months)

Conclusions

An accurate, precise, linear, and sensitive HPLC method was developed and validated for the analyses of nepicastat. The method has been used to quantitatively assay the API and bulk stability samples. A separate gradient extension of the assay method was developed, qualified and used for the analysis of nepicastat related organic impurities.

Acknowledgement

This work was funded by NIDA Contract No. N01DA-9-8890. Nepicastat was provided to NIDA through a Collaborative Research and Development Agreement between NIDA and Biotie Therapies.