Development and Validation of an HPLC-UV Method for 5-LIO(Me-3,2-HOPO), a Potent Actinide Chelator for Decorporation of Radionuclides in Personnel Exposed to Radiation

J. W. Hines[1], P. D. Elkins[1], D. P. Coleman[1], K. N. Raymond[2], R. J. Abergel[2], D. K. Shuh[2], P. S. Liu[3]
[1]Center for Analytical Chemistry and Pharmaceutics, RTI International, 3040 Cornwallis Road, Research Triangle Park, NC 27709
[2]Division of Chemical Sciences, Lawrence Berkeley National Laboratory, 1 Cyclotron Road, Berkeley, CA 94720
[3]Pharmaceutical Resources Branch, DCTD, National Cancer Institute, NIH, Bethesda, MD 20892

Purpose

5-LIO(Me-3,2-HOPO) is a potent tetradentate chelating ligand being developed as a decorporation drug for individuals internally contaminated with radionuclides. The compound has high affinity for actinides by design and, to a lesser extent, for iron. Development and validation of an HPLC method for quantitative assay and impurity profiling was undertaken. The strategy to derivatize the target ligand in situ, by addition of excess Fe(III) chloride, proved to be successful.

5-LIO-Me-3,2-HOPO
$C_{18}H_{22}N_4O_7$
Nominal Mol. Wt. 406.40

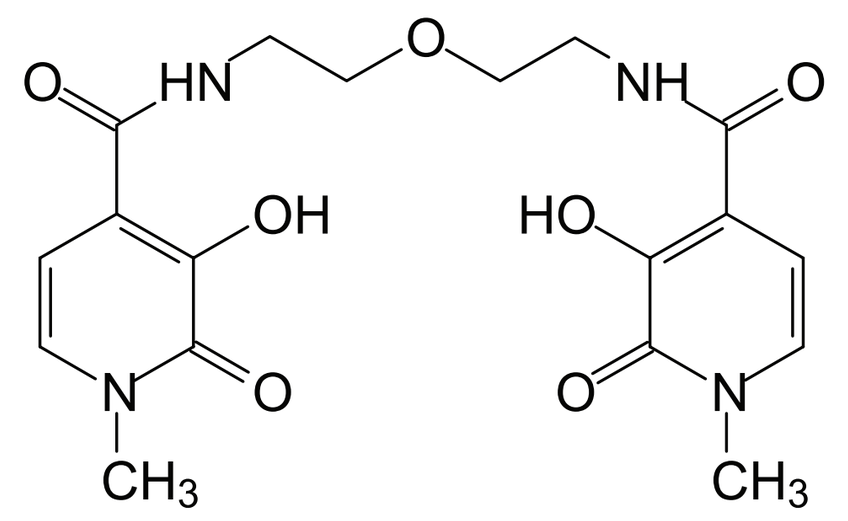

Methods

Test samples were prepared at 0.25 mg/mL in aqueous 0.4 mg/mL $FeCl_3$. Sample assay used an isocratic reverse phase HPLC-UV system in 12% acetonitrile/88% water/0.1% TFA. The HPLC impurity profile used gradient HPLC in 5-95% acetonitrile/water/~0.1% TFA. A Phenomenex Luna C18(2) column (150 mm X 4.6 mm, 3 µm) was used at 30˚C with 250 nm UV detection.

HPLC Assay Method Conditions:

Equipment:	HPLC systems (Waters) equipped with an autosampler, column-temperature-controller, photodioade array detector and Waters chromatographic data system
Mode:	Reverse phase, isocratic
Column:	Phenomenex Luna C18(2) column, 150 mm X 4.6 mm, 3 µm
Column Temp.:	30°C
Mobile Phase:	12% acetonitrile/88% water/0.1% TFA (v/v/v)
Flow Rate:	1.0 mL/minute
Detection:	UV at 250 nm
Sample Diluent:	0.4 mg/mL solution of $FeCl_3$ in DI water (fresh weekly)
Injection Volume:	10 µL
Run Time:	25 minutes
Assay Concentration:	0.25 mg/mL (nominal)

Representative HPLC Chromatograms for the Quantitative Analysis of 5-LIO-Me-3,2-HOPO Test Samples

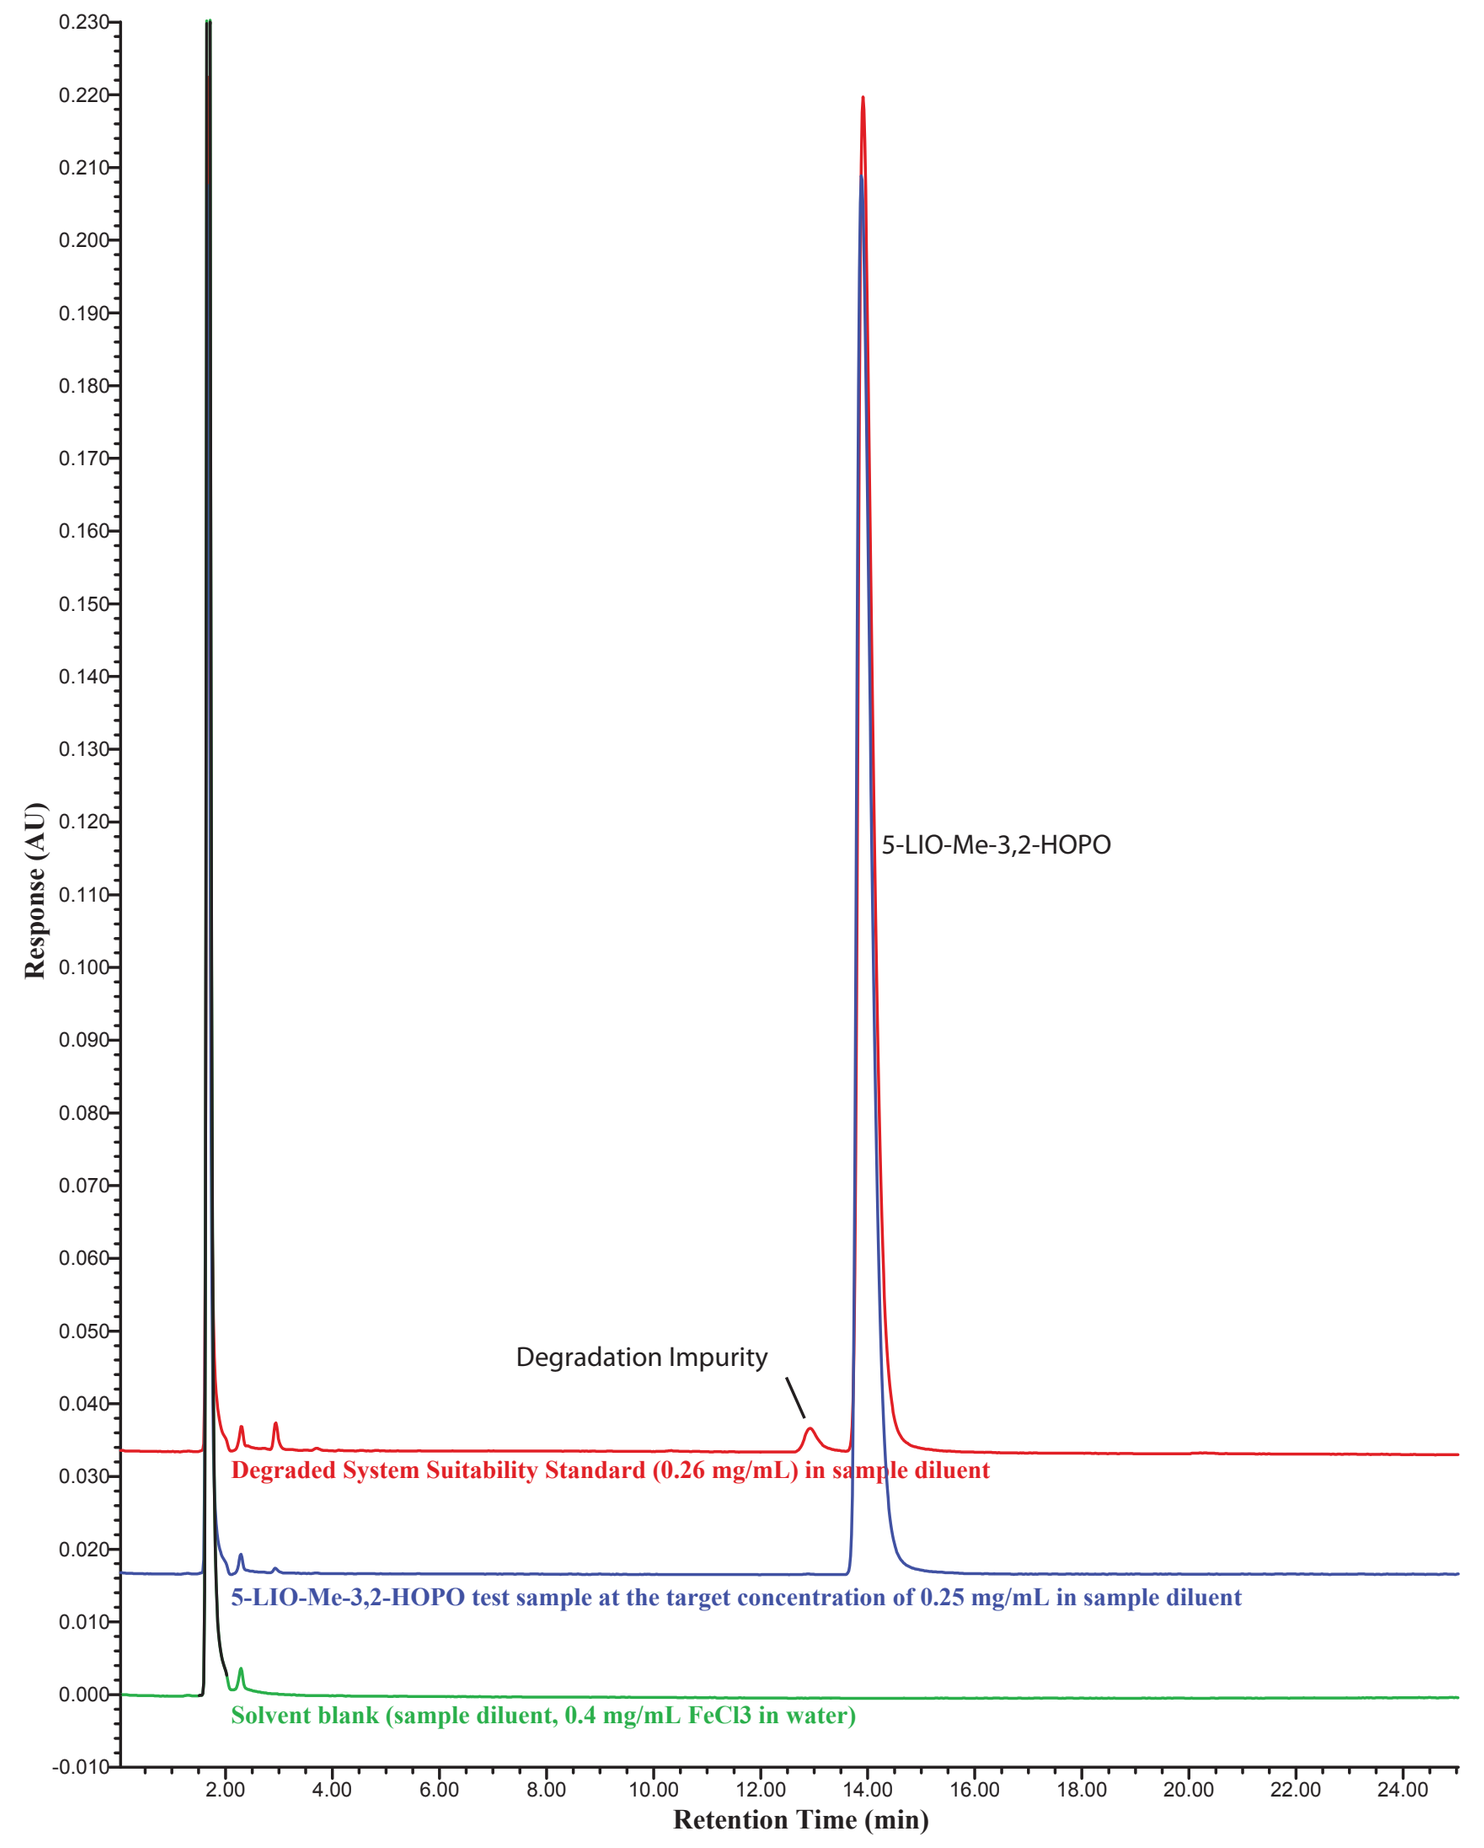
5-LIO-Me-3,2-HOPO
Degradation Impurity
Degraded System Suitability Standard (0.26 mg/mL) in sample diluent
5-LIO-Me-3,2-HOPO test sample at the target concentration of 0.25 mg/mL in sample diluent
Solvent blank (sample diluent, 0.4 mg/mL FeCl3 in water)
Response (AU)
Retention Time (min)

Results

Samples were prepared at a 4:1 molar ratio of $FeCl_3$ to fully chelate drug during chromatography. Linearity was demonstrated over the range 0.05-0.39 mg/mL, with a correlation coefficient of ≥ 0.9999 and y-intercept -0.5% of the nominal assay level. Method precision for multiple prepared samples was 0.4% (RSD) for response and 0.1% for retention with accuracy (recovery) of 100.0-100.4% over the linearity range. Ruggedness was evaluated across two days, two analysts, and two HPLC systems. Robustness was demonstrated for method performance with changes in retention, column temperature, TFA concentration, and sample concentration. Sample solution stability was confirmed up to 4 days at refrigerator and room temperatures. Specificity was demonstrated by resolution of impurities and forced decomposition products using photodiode array peak purity.

Linearity:

Results for Determination of Assay Linearity for 5-LIO-Me-3,2-HOPO

Assay Level (%)	Actual Conc. (mg/mL)	Analyte Response (area)	Calc Conc. (mg/mL)	% Error
20	0.0516	727800	0.0510	-1.2
50	0.130	1907095	0.131	1.0
80	0.209	3054548	0.210	0.3
100	0.261	3776554	0.259	-0.8
120	0.313	4579374	0.314	0.2
150	0.391	5721260	0.391	0.1

Concentration Calculated from regression line: Y = a + bX
a = -19836, b = 14669258
r^2 = 0.999897, r = 0.999948
rss = 1687485160
Intercept % = -0.5

All acceptance criteria were met as follows: (1) overall linearity ≥99.8% (r^2) of total variance explained by the regression line over the specified linear range, (2) absolute intercept ≤ 2.0% of the response at the assay concentration, and (3) the % Error was ≤ 10% over the range 20-150% and ≤ 2.5% over 80-120%.

Linearity for HPLC/UV Detection of 5-LIO-Me-3,2-HOPO over the Assay Concentration Range 0.05–0.39 mg/mL

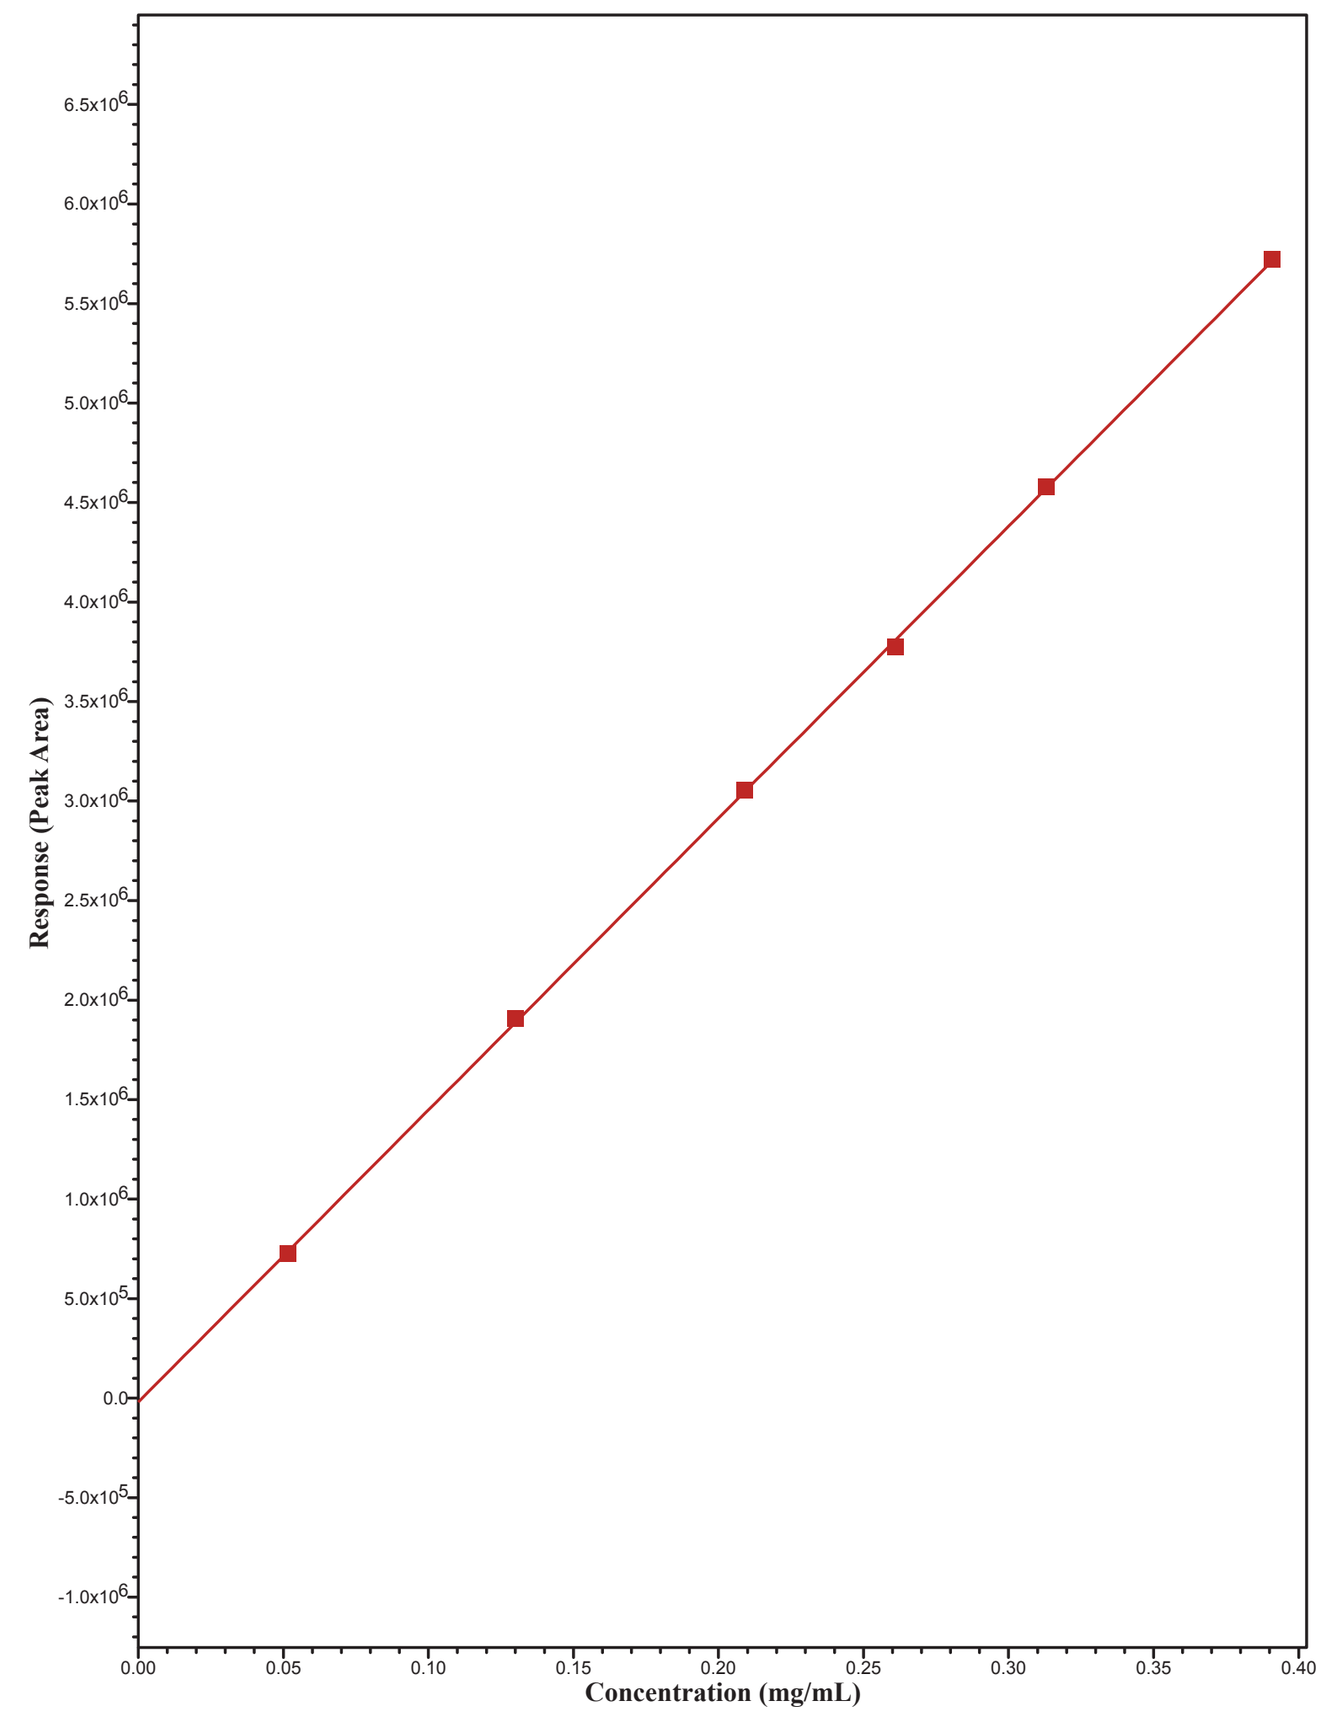
Response (Peak Area)
Concentration (mg/mL)

Precision (Repeatability):

Method repeatability was determined from analysis of nine standard solutions containing 5-LIO-Me-3,2-HOPO, individually prepared, covering the range of the procedure (3 concentrations/3 replicates each). The concentration levels evaluated were at 20%, 100% and 150% of the nominal drug substance assay concentration of 0.25 mg/mL. Precision acceptance criteria with respect to chromatographic retention (%RSD ≤ 1.0 for k') and RF (%RSD ≤ 2.0) were met.

Results for Determination of Method Repeatability for 5-LIO-Me-3,2-HOPO Drug Substance Assay Method

Assay Level (%)	k'		RF[a]	
	Mean (n=3)	%RSD	Mean (n=3)	%RSD
20	4.1[b]	0.0	6.96×10^{-8}	0.4
100	4.1	0.1	6.83×10^{-8}	0.4
150	4.1	0.1	6.84×10^{-8}	0.2

[a]Analyte concentration/analyte response
[b]Original k' range was 3-5 but was increased to provide impurity resolution

Accuracy:

Assay accuracy was determined by quantitation of the assay precision samples using the calibration curve calculated from the linearity solutions. Accuracy was measured as the deviation of test results from actual values.

Results for Determination of Accuracy for 5-LIO-Me-3,2-HOPO Drug Substance Assay Method

Assay Level (%)	Replicate	Actual Conc. (mg/mL)	Calc. Conc. (mg/mL)	Mean Recovery (%)
20	1	0.0542	0.0540	100.4
	2	0.0538	0.0540	
	3	0.0465	0.0470	
100	1	0.264	0.264	100.3
	2	0.241	0.241	
	3	0.230	0.232	
150	1	0.391	0.391	100.0
	2	0.348	0.349	
	3	0.372	0.371	

The accuracy at all three assay concentrations met acceptance criteria for recovery at the assay concentration (98.0-102.0%) and at the high and low levels for the range (97.0-103.0%).

Ruggedness (Intermediate Precision):

Method ruggedness was evaluated by two different analysts on two different analytical columns and HPLC systems, and on a different day to establish the effects of typical variations on the precision of the analytical procedure. The ruggedness (as %RSD) obtained for the analyte chromatographic retention and analyte response factor (RF) were found to be within the acceptable criteria of %RSD ≤ 1 for retention at the target assay concentration and %RSD ≤ 2 for RF for the assay.

Results for Determination of Method Ruggedness for 5-LIO-Me-3,2-HOPO Drug Substance Assay Method

Assay Level (%)	k'		RF[a]	
	Average (n=3)	%RSD	Average (n=3)	%RSD
20	9.3	0.10	6.72×10^{-8}	1.4
100	9.1	0.02	6.84×10^{-8}	0.5
150	9.0	0.03	6.87×10^{-8}	0.1

[a]Analyte concentration/analyte response

Robustness:

The robustness of the 5-LIO-Me-3,2-HOPO method was determined by examining the effect of slight variations on the method performance; specifically, the effect of changes in column temperature, the capacity factor (k'), the analyte concentration, and the TFA concentration in the mobile phase. Although analyte concentration was also evaluated in the assay linearity, it was evaluated in robustness in terms of possible unintended effects on the use of $FeCl_3$ diluent in sample preparation. Robustness was assessed by evaluation of system suitability. Results from all robustness tests met all system suitability criteria.

Results for Determination of Method Robustness for 5-LIO-Me-3,2-HOPO Drug Substance Assay Method

Testing		Results				
Parameter	Values	k'	N	T	R[a]	% RSD for RF
Capacity Factor[b]	~6	5.97	11460	1.78	1.35	0.2
	~9	8.43	11657	1.91	2.48	0.1
Column Temperature	25˚C	4.84	11255	1.65	---[c]	0.1
	35˚C	3.86	12448	1.56	---	0.2
Concentration of TFA in the mobile phase	0.09%	4.00	10655	1.67	---	0.3
	0.11%	4.16	12328	1.57	---	0.1
Concentration of Analyte (mg/mL)	0.22	4.26	12164	1.58	---	0.1
	0.28	4.26	11207	1.69	---	0.1

System Suitability Acceptance criteria:
k' = 6-9[b]
N = >5,000
T = 0.5 – 2.0
Resolution = ≥1.0
%RSD for RF = ≤2.0% (n=5)

[a]*Resolution* of impurity from the analyte peak
[b]Recommended retention range to ensure resolution of a degradation impurity (see assay figure), other values follow the initial criteria of k' = 3-5 and were not repeated
[c]Not measured

Specificity:

Assay specificity demonstrates the lack of interferences which could affect the accuracy of the analytical method (i.e., measurement of the analyte peak). For the 5-LIO-Me-3,2-HOPO assay method, accelerated degradation studies were used to demonstrate specificity in the presence of all possible impurities including those from degradation of the drug substance. This evaluation was performed using photodiode array (PDA) detection and the manufacturer's algorithms for peak purity to verify spectral homogeneity and library matching to confirm the identity of the analyte peak.
Samples of 5-LIO-Me-3,2-HOPO for assay specificity evaluation were prepared and stored under conditions of forced degradation including dry, light, moisture, elevated temperature, acid, base, and oxidation. After the storage period, samples were diluted for analysis to a nominal concentration of 0.25 mg/mL 5-LIO-Me-3,2-HOPO in 20-mL volumetric flasks. Controls containing no analyte were also exposed with the samples for assessment of chromatographic background.

Specificity of 5-LIO-Me-3,2-HOPO Drug Substance Assay Method

Storage Temperature	Light Exposure	Storage Solvent[a]	Storage Duration (h)	% Remaining
85˚C	dark	dry (as is)	24	99.8
85˚C	dark	H_2O	24	96.2
ambient	light	H_2O	24	99.9
ambient	dark	0.1 N NaOH[b]	24	97.6
ambient	dark	0.1 N HCl	24	100.0
ambient	dark	0.006% NaClO	1	93.8

[a]2 mL of each storage solvent were added to 5 mg (± 20%) of test sample.
[b]After storage, 0.2 mL of 1N HCl was added prior to diluting to 20 mL.

Although 3% H_2O_2 is normally used for the oxidative degradation, initial evaluation of the condition showed that test sample disappearance was nearly instantaneous requiring a milder oxidation reagent (i.e., NaClO) and shorter storage time to allow some undegraded material to remain at the end of the storage period. All test samples met specificity requirements of "Purity Angle < Purity Threshold" for chromatographic peak homogeneity, and "Match Angle < Match Threshold" for peak ID as 5 LIO-Me-3,2-HOPO for the presence of degradation products under the stated assay conditions.

Solution Stability:

The stability of the 5-LIO-Me-3,2-HOPO drug substance in sample solutions prepared for HPLC assay was evaluated up to 4-days (96 h) by setting aside an aliquot of the sample solution under ambient laboratory conditions of temperature, humidity, and light (overhead fluorescent) and a second aliquot stored refrigerated (2-8 °C) in the dark.
After storage for 4 days, an aliquot from each of the two storage locations was analyzed (triplicate injections) vs. a fresh sample (standard), and the % recovery was calculated for each. The % recovery obtained for the solutions stored up to and including 4 days met acceptance criteria (% Recovery = 98-102%), and the overall impurity profile was not significantly changed during storage.

5-LIO-Me-3,2-HOPO Related Impurities:

5-LIO-Me-3,2-HOPO related organic impurities are determined by a separate gradient extension of the 5-LIO-Me-3,2-HOPO assay method. This method was qualified by demonstrating linearity over a range of 0.050 to 0.378 mg/mL (r^2 = 0.99938) and establishing a detection limit of 0.1% assay concentration with a S/N of 16:1.

HPLC Impurities Method Conditions:

Column:	Phenomenex Luna C18(2) column, 150 mm X 4.6 mm, 3 µm
Column temperature:	30°C
Detector:	UV at 250 nm
Flow rate:	0.9 mL/minute
Injection volume:	10 µL
Diluent:	$FeCl_3$ in deionized water, 0.4 mg/mL
Mobile phase A:	5% acetonitrile/95% water/0.1% TFA (v/v/v)
Mobile phase B:	95%acetonitrile/5% water/0.08 % TFA (v/v/v)
Gradient Program:	See below
Sample:	~0.25 mg/mL in sample diluent
Standard:	~0.25 mg/mL in sample diluent

Time	Mobile Phase A (%)	Mobile Phase B (%)
0.0	100	0
5.0	100	0
35.0	0	100
36.0	100	0
47.0	100	0

System Suitability for HPLC Impurity Profile Method

Parameter	Description	Acceptance Criteria
k'	Capacity factor for gradient elution	3-7
N	Column efficiency	≥ 10000 plates/column
T	Peak asymmetry	0.5 - 2.0

Representative 5-LIO-Me-3,2-HOPO Drug Substance Impurities Chromatogram

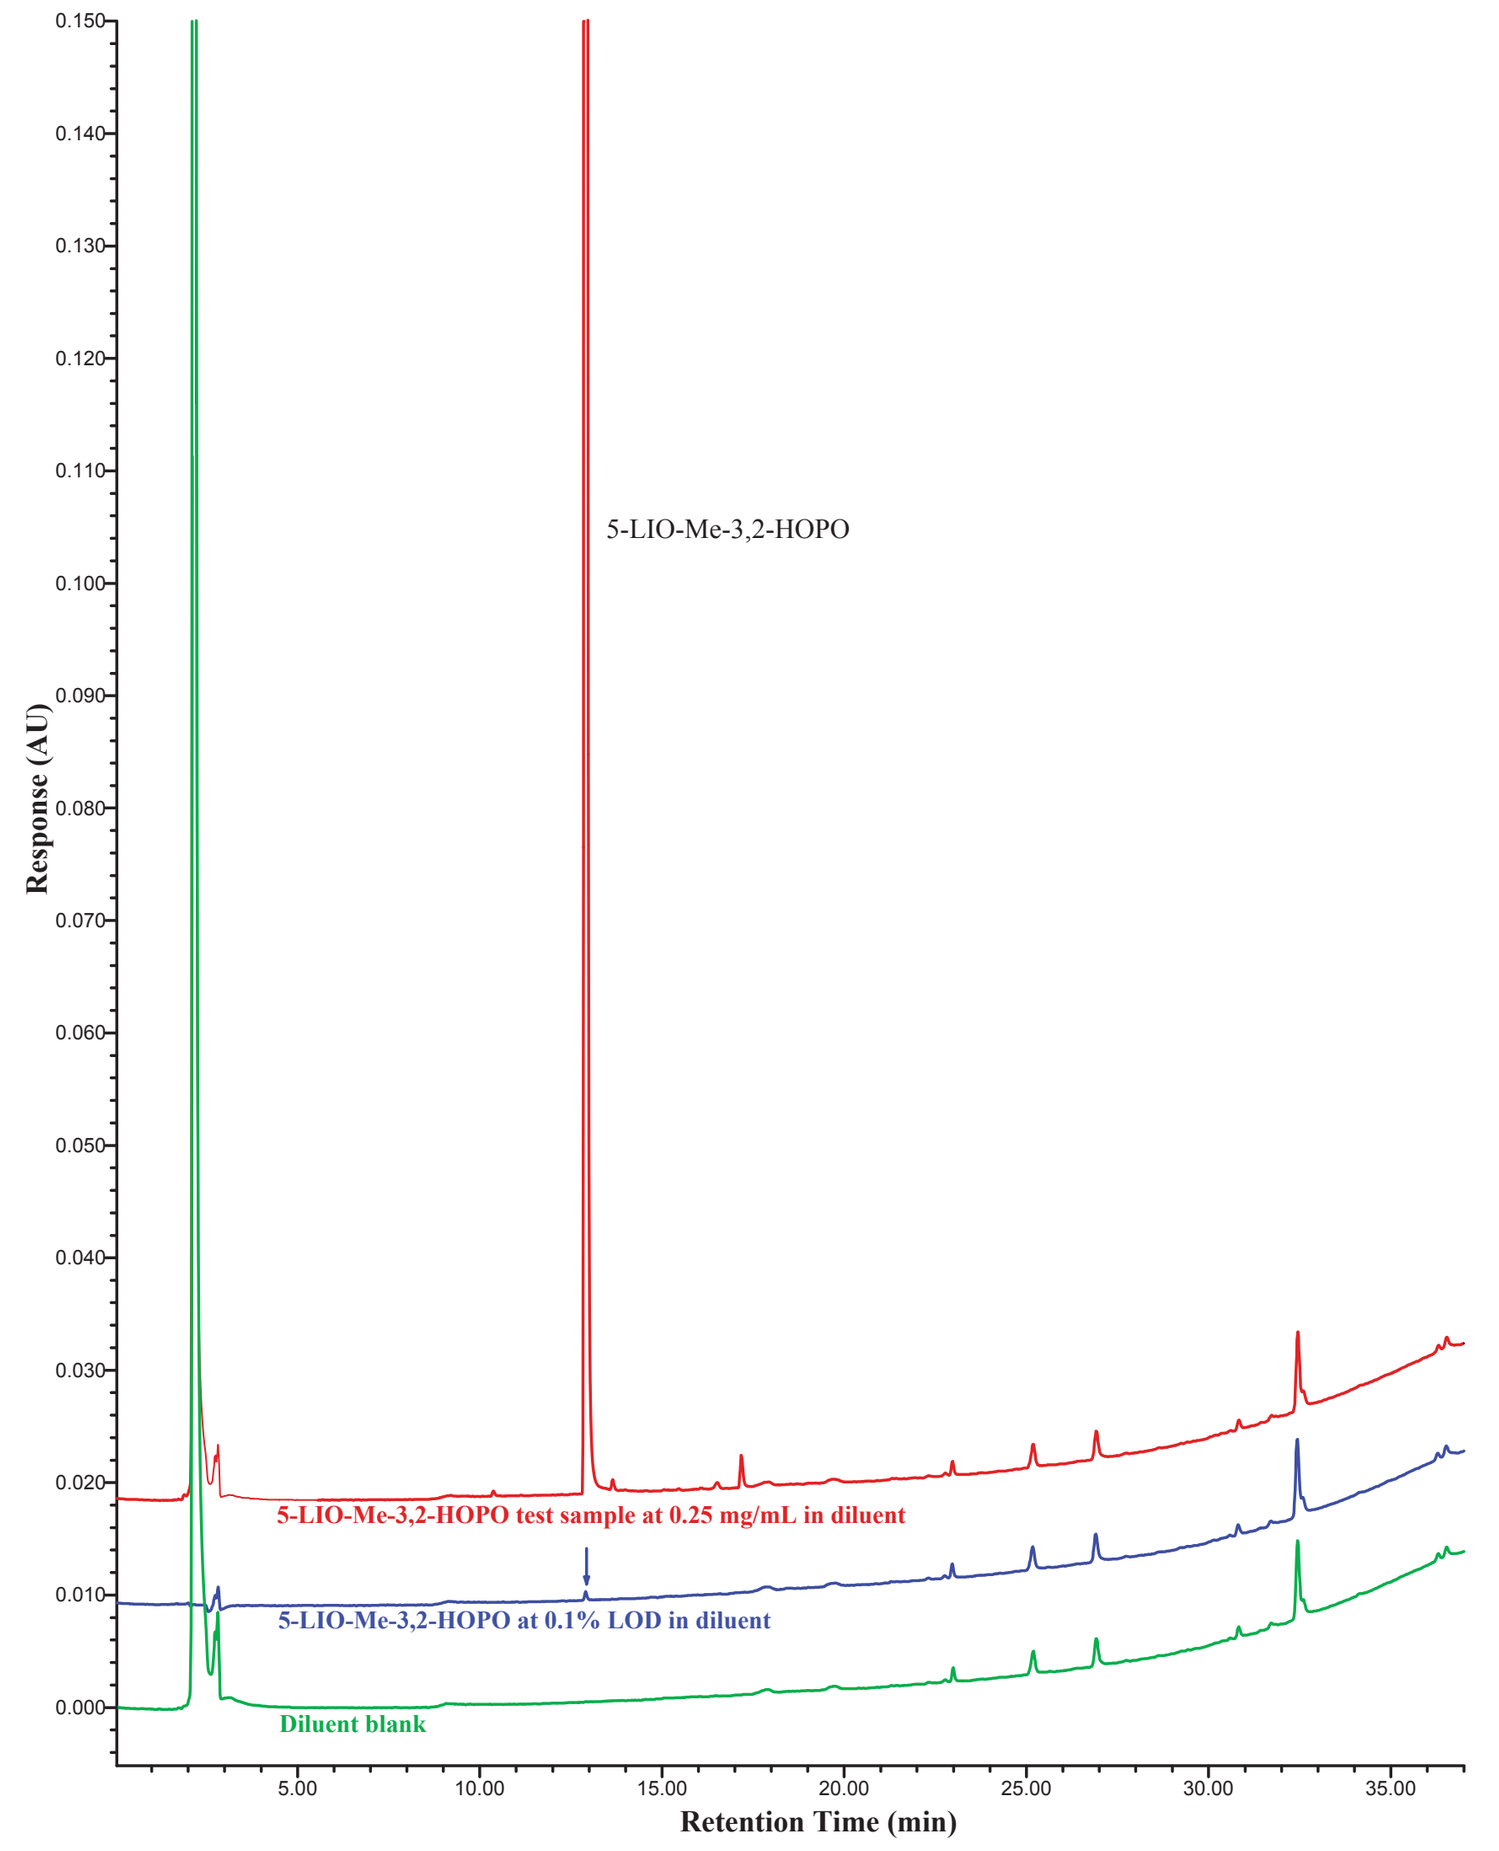
5-LIO-Me-3,2-HOPO
5-LIO-Me-3,2-HOPO test sample at 0.25 mg/mL in diluent
5-LIO-Me-3,2-HOPO at 0.1% LOD in diluent
Diluent blank
Response (AU)
Retention Time (min)

5-LIO-Me-3,2-HOPO Drug Substance Stability:

The described methods for analysis of 5-LIO-Me-3,2-HOPO and related impurities have been used successfully at RTI to evaluate the stability of a cGMP batch of 5-LIO-Me-3,2-HOPO drug substance under ICH conditions of heat and humidity for drug substances including accelerated (40°C/75% RH), intermediate (25°C/60% RH), and long-term (5°C) stability evaluations.

Trend Chart for Assay of 5-LIO-Me-3,2-HOPO Drug Substance Under ICH Stability

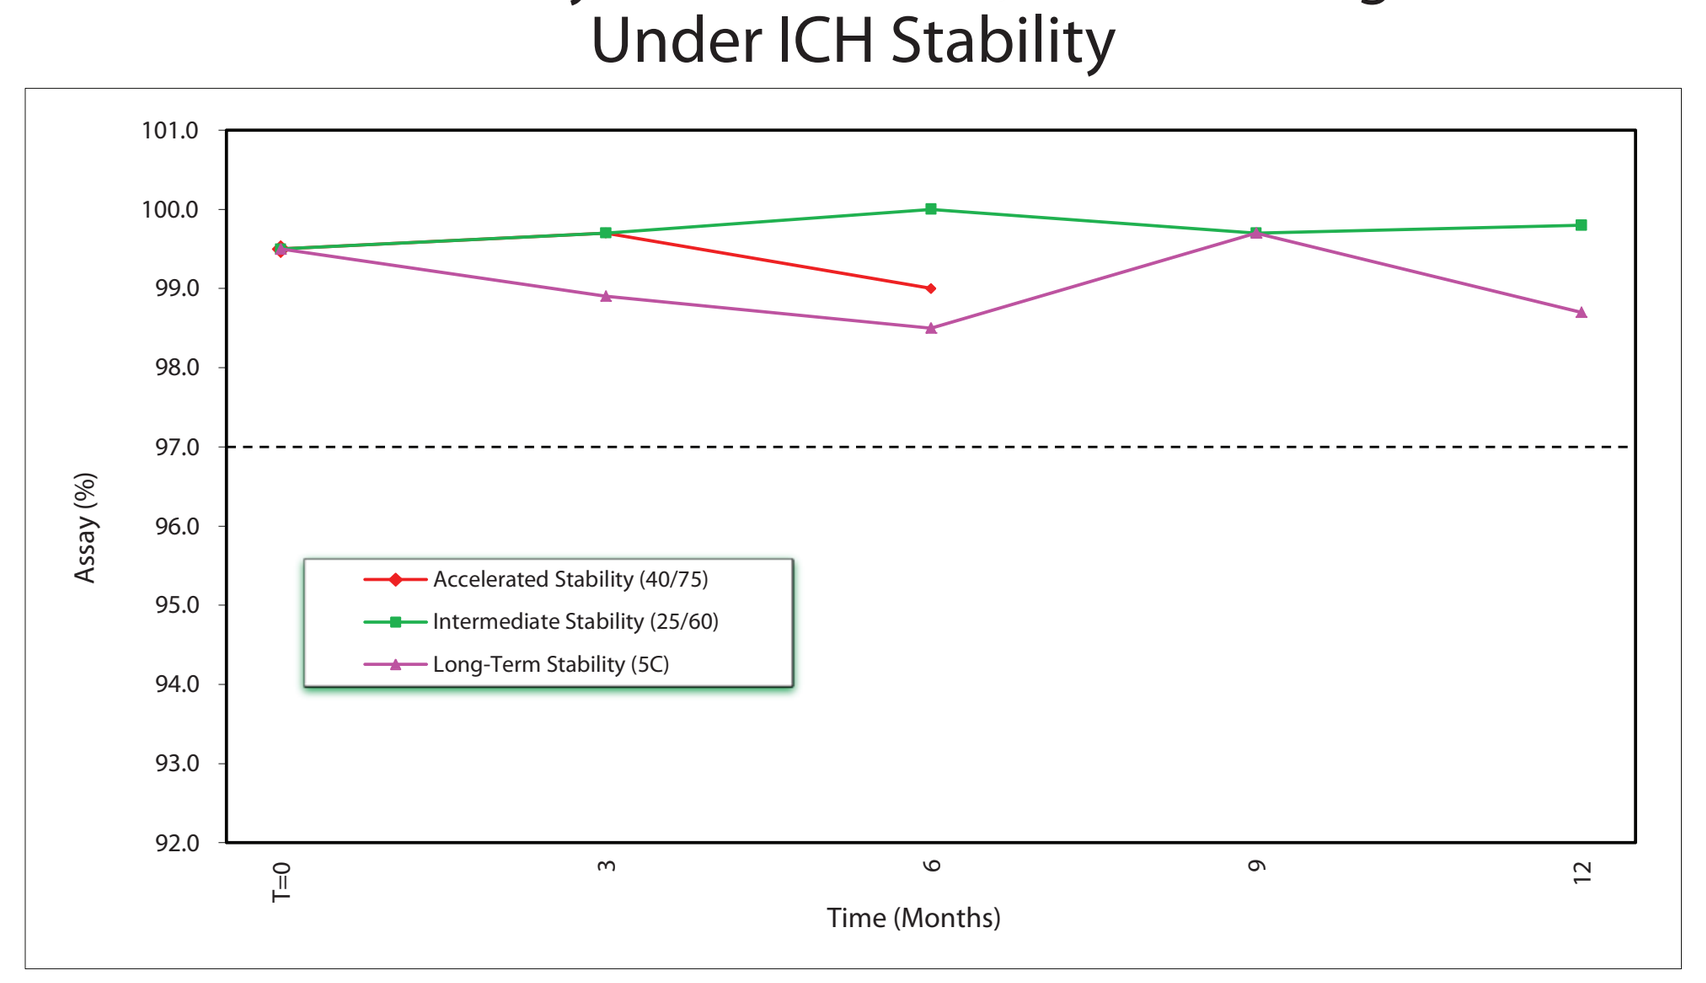
Accelerated Stability (40/75)
Intermediate Stability (25/60)
Long-Term Stability (5C)
Assay (%)
Time (Months)

Conclusions

An accurate, precise, linear, and sensitive HPLC assay method was developed and validated for analysis of the compound. The method was used to quantitatively evaluate the API and drug substance stability. A gradient extension of the assay method was developed, qualified and used for analysis of drug-related organic impurities.

Acknowledgement

This work was supported by the NIH Common Fund and NIAID under NCI Contract No. N02-CM-72202.